Abstract #: Machine learning and statistical prediction of overall survival (OS) from pre-dose plasma biomarkers in a randomized phase 2 trial (1801 Part 3B) of the GSK-3 inhibitor elraglusib with gemcitabine/nab-paclitaxel (GnP) in metastatic pancreatic ductal adenocarcinoma (mPDAC): application toward patient enrichment

Taylor Weiskittel, Austin Koukol, Caroline Kellinger, Andrey Ugolkov, Chris Seifarth, Hu Li, Devalingam Mahalingam, Leiqing Zhang, Benedito Carneiro, Wafik El- Deiry, Andrew Mazar

ACTUATE THERAPEUTICS · BROWN · MAYO CLINIC

Background

Preliminary results of the 1801 Part 3B trial (NCT03678883) showed statistically significant benefits for elraglusib+GnP versus GnP for 1-year survival and mOS in mPDAC (companion abstract: Mahalingam et al.). We investigated plasma levels of cytokines/ chemokines/ soluble cell receptors/growth factors (CCSG) as potential biomarkers of favorable outcomes on elraglusib.

Updated with Topline Data from April 2025

Figure 1: Overall Survival for the 1801 Study

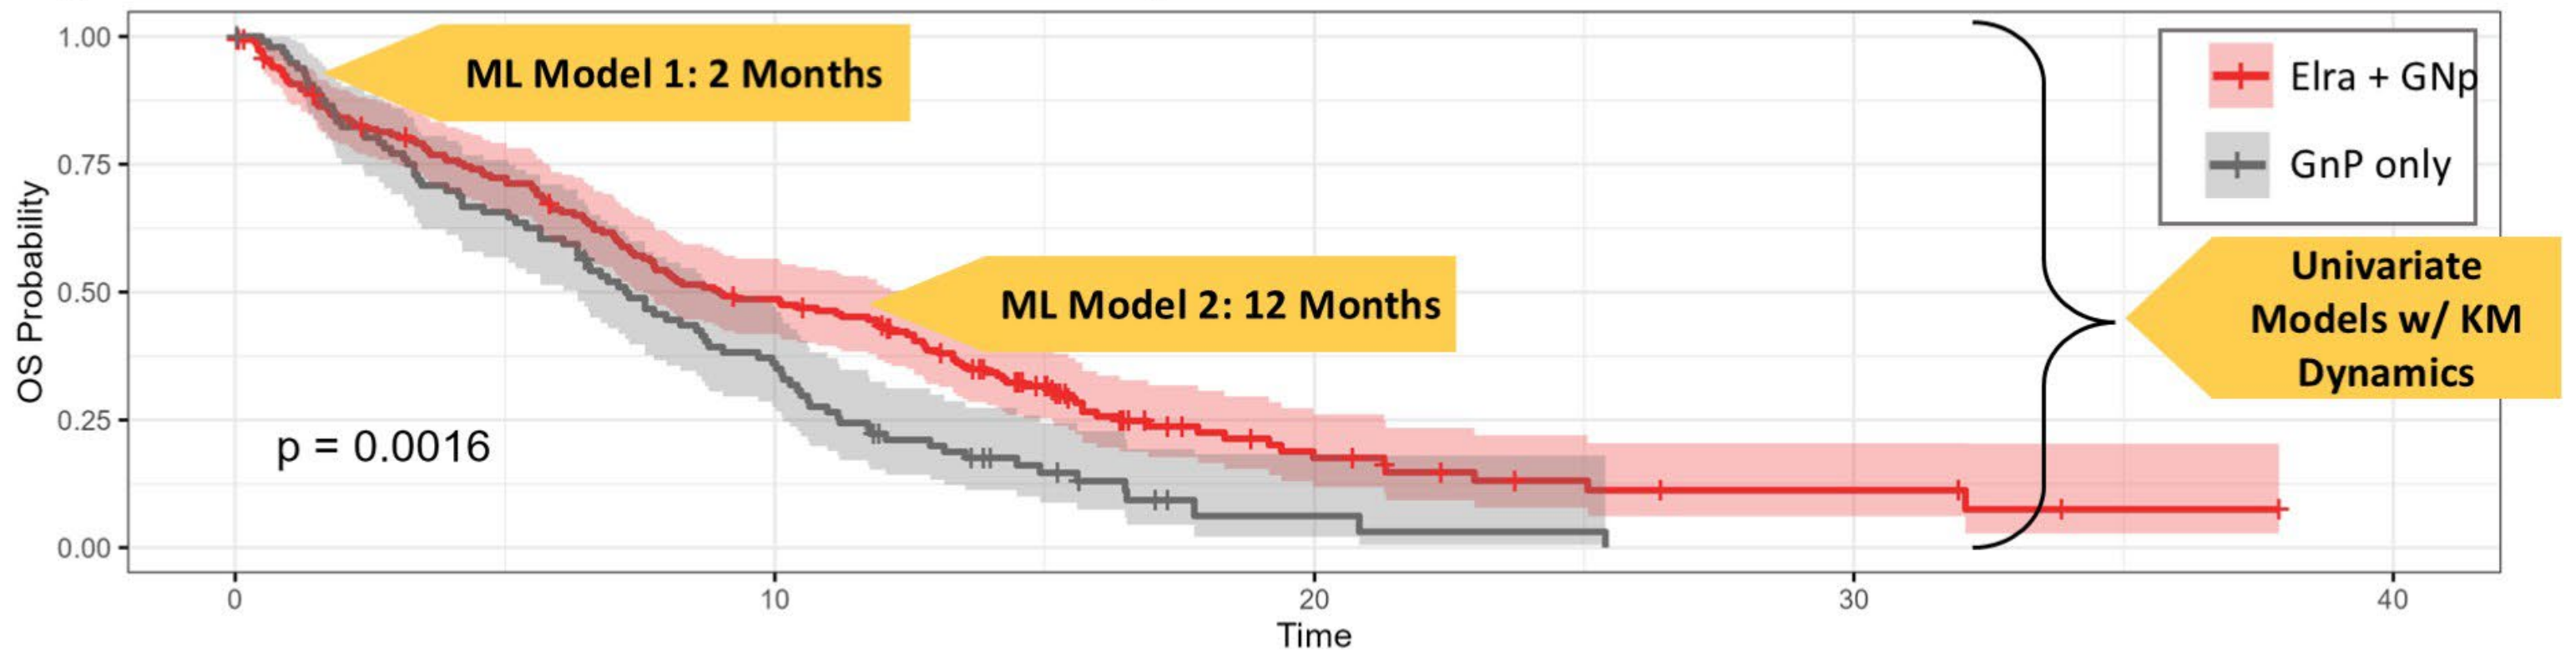
ML Model 1: 2 Months
ML Model 2: 12 Months
Univariate Models w/ KM Dynamics
Elra + GNp
GnP only
p = 0.0016
OS Probability
Time

Machine Learning Pipeline and Results

40 CCSGs measured in Pre-Dose Plasma n=224

Remove Patients LTFU before 2 mo. n=221

Binarize as Alive/ Dead at 12 mo. Alive=190 Dead=31

5xCV Screen Unoptimized Machine Learning Architectures → 5xCV Top Architectures Hyperparameter Grid Search → 5xCV Evaluate Final Algorithms

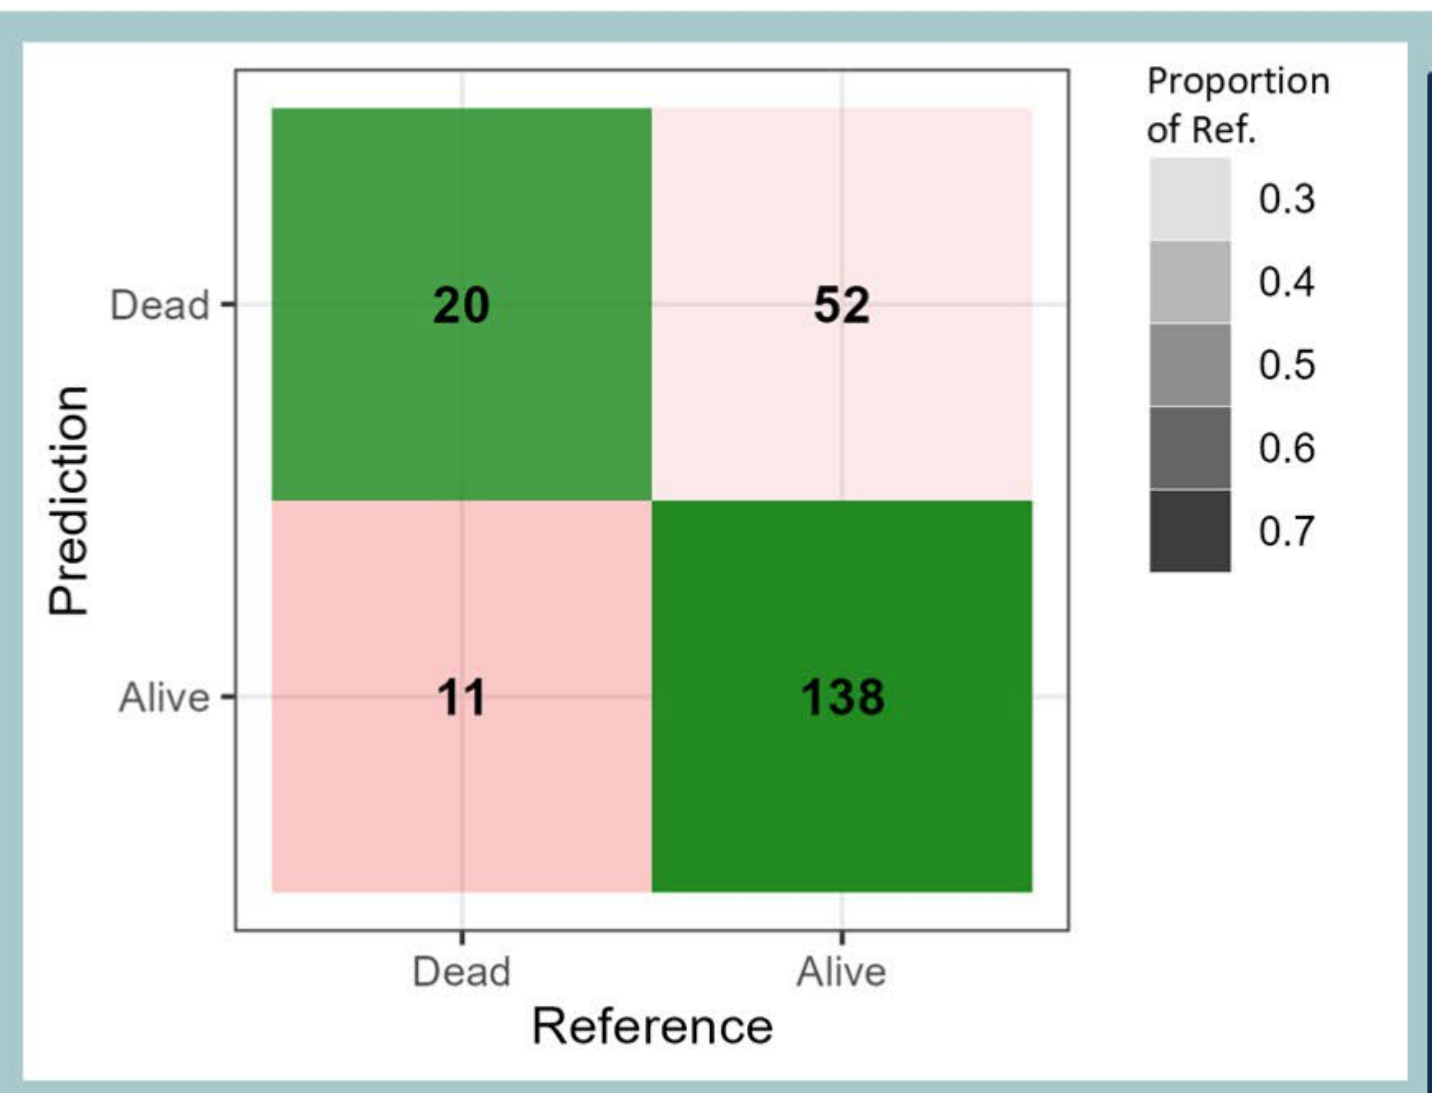
Proportion of Ref.
Dead
Alive
20
52
11
138
Prediction
Reference

Linear Support Vector Classifier

Mean (SD)

Accuracy: 0.765 (0.033)

Balanced Accuracy: 0.705 (0.101)

40 CCSGs measured in Pre-Dose Plasma Elra + GnP n=148

Remove Patients LTFU before 2 mo. n=148

Binarize as Alive/ Dead at 12 mo. Alive=62 Dead=86

5xCV Screen Unoptimized Machine Learning Architectures → 5xCV Top Architectures Hyperparameter Grid Search → 5xCV Evaluate Final Algorithms

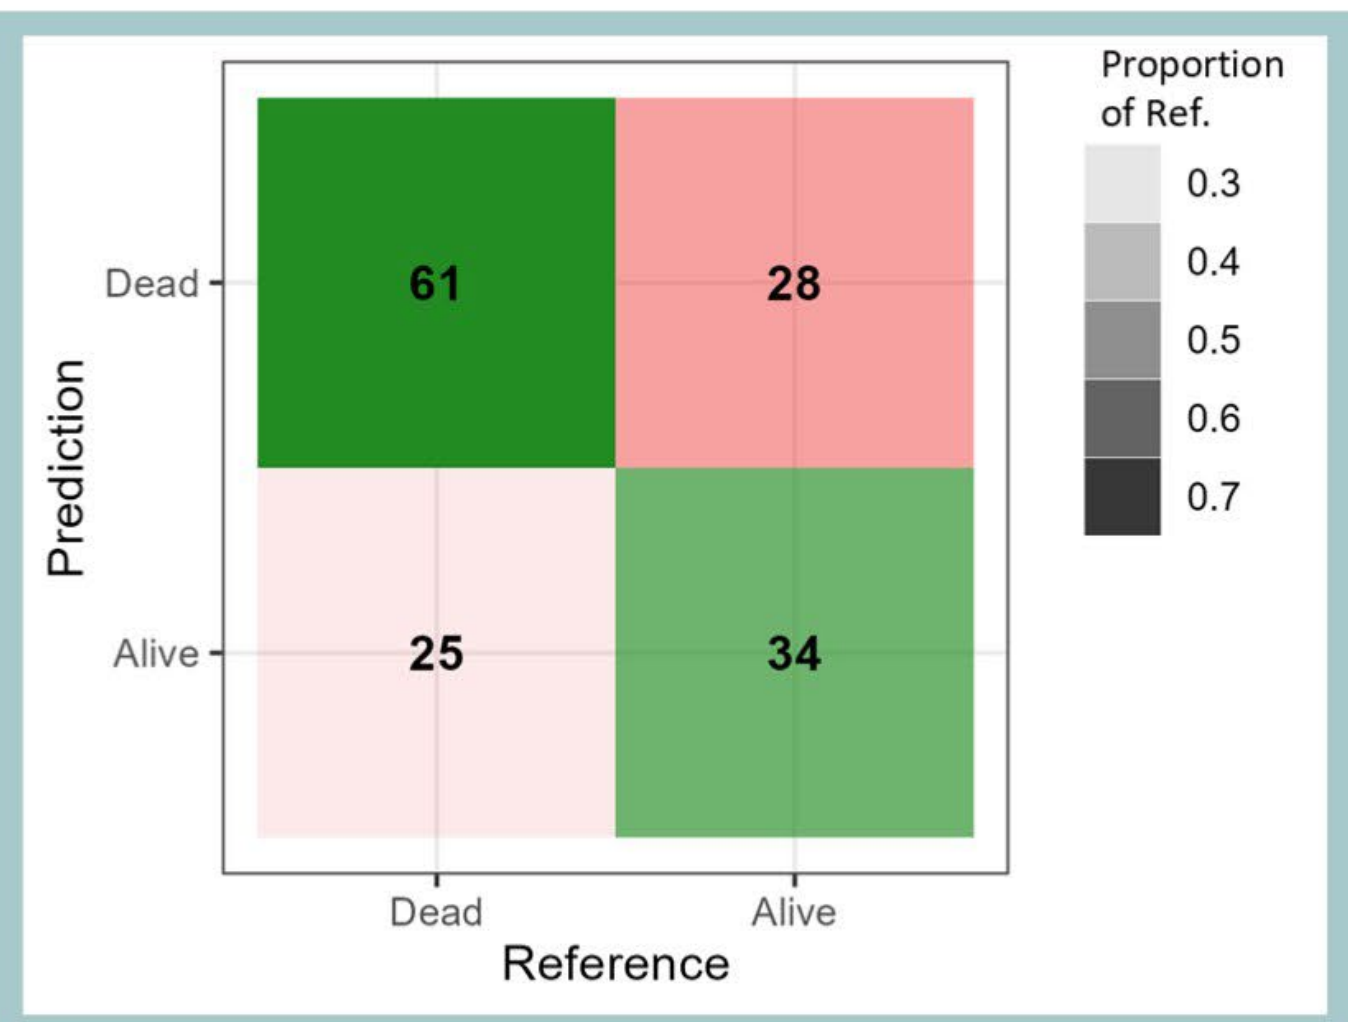
Proportion of Ref.
Dead
Alive
61
28
25
34
Prediction
Reference

Linear Support Vector Classifier

Mean (SD)

Accuracy: 0.639 (0.082)

Balanced Accuracy: 0.633 (0.085)

Univariate Pipeline and Results

40 CCSGs measured in Pre-Dose Plasma

Elra + GnP = 155 patients → Single CCSG Optimized KM Cutoffs on Elra + GnP → 21 Stat. Sig. OS Markers → 5-Fold Cross Validation → 18 Stat. Sig. OS Biomarkers on 5xCV

GnP only = 76 patients → Apply Elra + GnP Cutoffs to GnP only → 20 Stat. Sig. GnP Stratification *Markers of Overall Prognosis* → Single CCSG Optimized KM Cutoffs on GnP only → *13 Stat. Sig. OS Biomarkers Potential Markers of Overall Prognosis AND/OR Elraglusib Response*

→ *7 Uniquely Significant Elraglusib Markers* → *1 with Trends Opposite Control Survival Data*

Figure 2: Univariate Biomarkers with Unique Patient Selection Properties. A) CCL2 stratification of Elra + GnP patients compared to GnP only. B) TNFRSF6 stratification of Elra + GnP patients compared to GnP only.

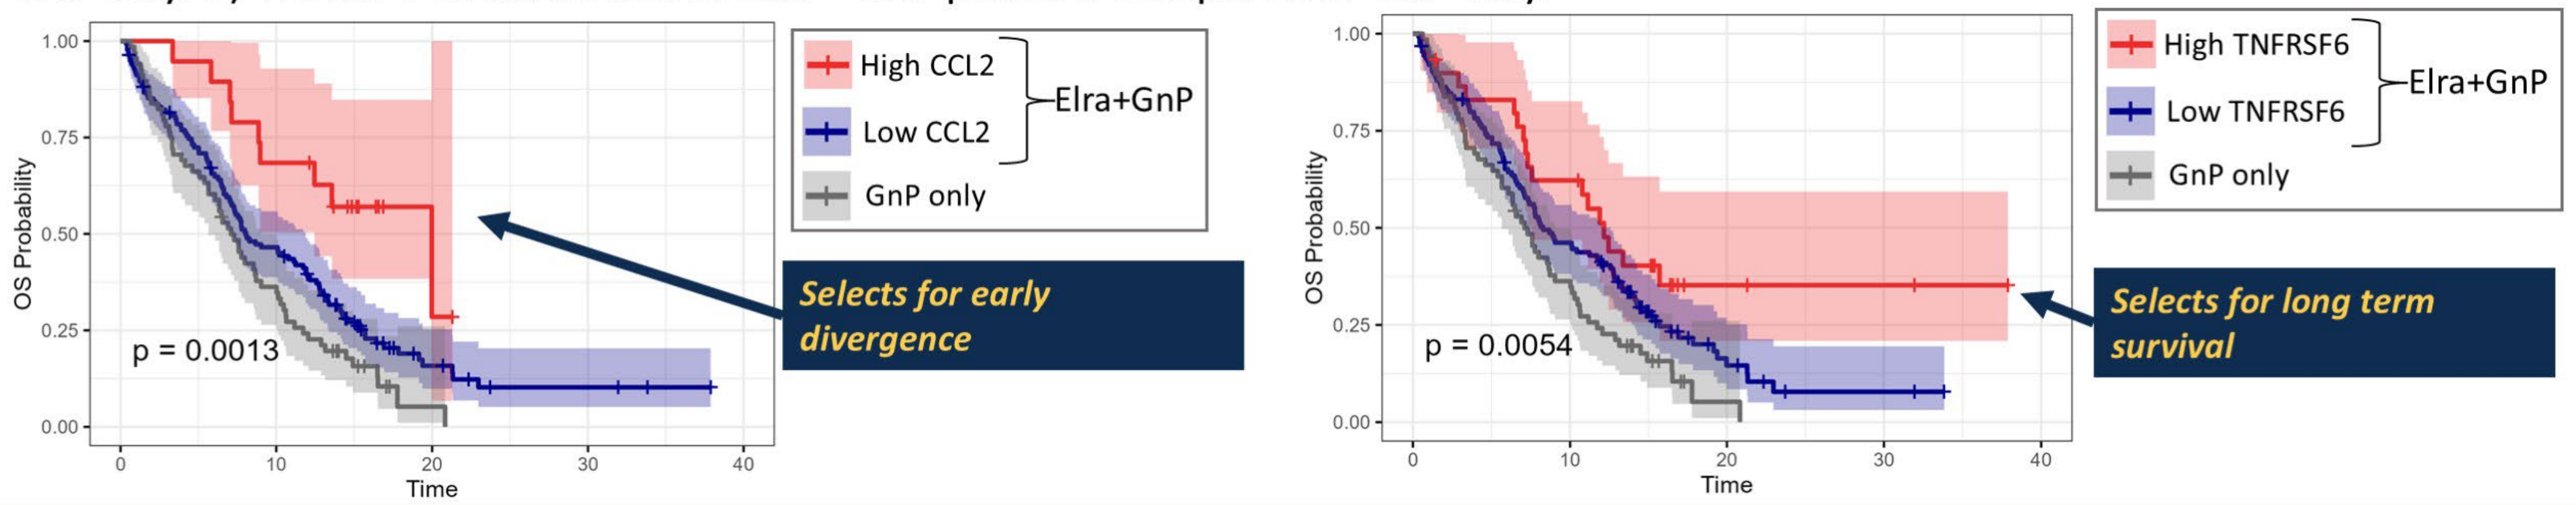
High CCL2
Low CCL2
Elra+GnP
GnP only
p = 0.0013
Selects for early divergence
High TNFRSF6
Low TNFRSF6
GnP only
p = 0.0054
Selects for long term survival
OS Probability
Time

Figure 3: CXCL2 as a Univariate Biomarker. A) CXCL2 stratification of Elra + GnP patients. B) 5xCV for CXCL2 stratification of Elra + GnP patients. C) CXCL2 stratification cutoff from A applied to GnP only patients. D) Elra + GnP patients stratified by CXCL2 compared to GA only patients. E) Number of patients included in the high and low cohorts using the CXCL2 stratification cutoffs for both trial arms.

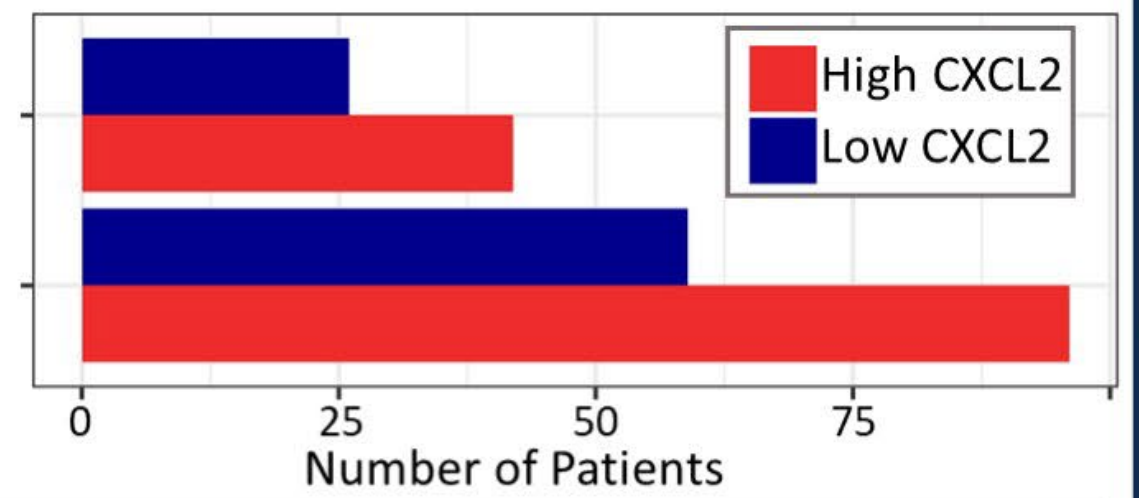
High CXCL2
Low CXCL2
Number of Patients

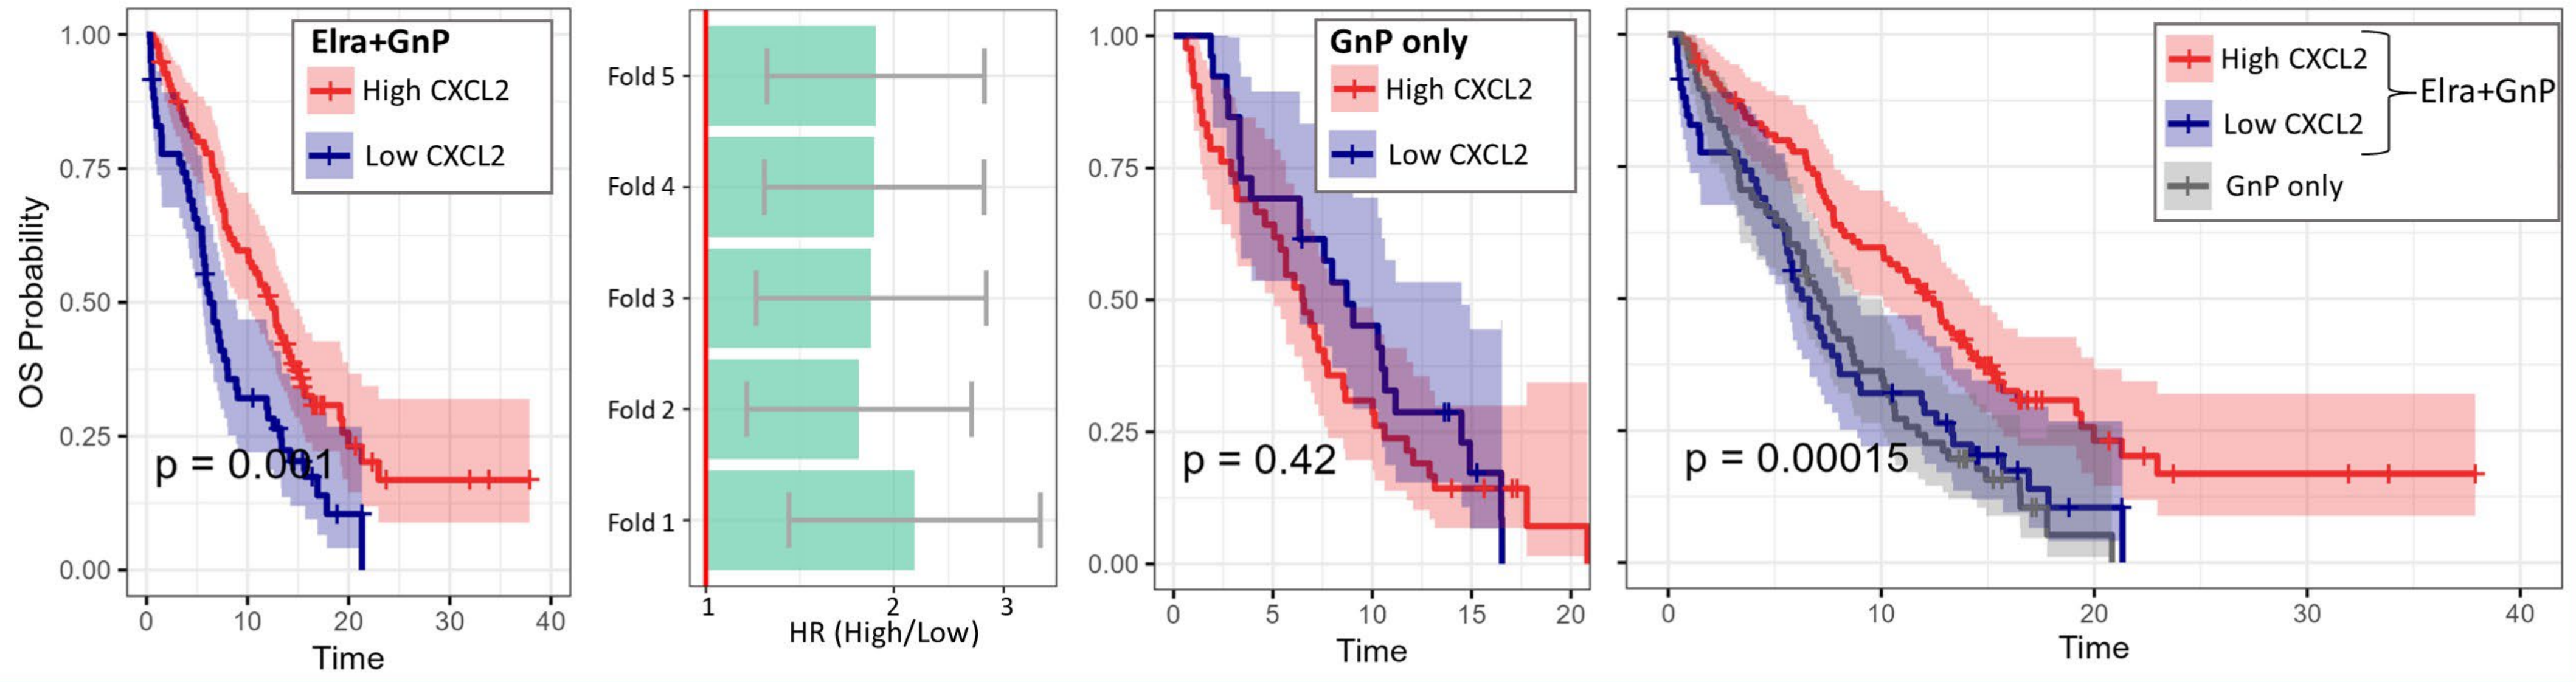
Elra+GnP
High CXCL2
Low CXCL2
p = 0.001
Fold 5
Fold 4
Fold 3
Fold 2
Fold 1
HR (High/Low)
GnP only
p = 0.42
GnP only
p = 0.00015
OS Probability
Time

	Elraglusib+Gnp			GnP only		
Cytokine	Cutpoint	P-value	HR	Cutpoint	P-value	HR
CCL3.MIP.1.alpha	483.74	p = 0.017	0.61	486.92	p = 0.061	0.58
CXCL2.Gro.beta.MIP.2.CINC.3	357.86	p = 0.001	1.85	826.65	p = 0.0063	0.41
IL.1.alpha.IL.1F1	16.62	p = 0.042	0.48	18.51	p = 0.082	1.99
IL.18.IL.1F4	153.35	p = 0.0055	0.59	264.98	p = 0.15	0.63
TGF.beta.1	10065.87	p = 0.033	0.67	2638.24	p = 0.075	0.41
TRAIL.R3.TNFRSF10C	4700.09	p = 0.045	0.68	2427.90	p = 0.29	0.69
TRAIL.TNFSF10	92.18	p = 0.0089	1.62	60.32	p = 0.22	1.56

Pre-dose plasma CCSG are promising predictive biomarkers of elraglusib clinical outcomes.

Future Directions

- Patient predose plasma collection on future trials will be tested for the seven cytokines
- Investigate sequential combinations of univariate CCSGs for optimal patient selection and stratification
- Further optimize multivariate machine learning models for increased accuracy in predicting 1-year OS
- Compare machine learning and sequential univariate stratification

Oral (Abstract 4006): Mahalingam et al.
Preliminary results from 1801 part 3B elraglusib in combination with GnP versus GnP with previously untreated mPDAC.
Session: Saturday, May 31, 2025, 3:00 PM-6:00 PM CDT. Hall D1.