Targeting GSK-3: a novel approach to enhance glioblastoma chemosensitivity

Ugolkov A,[1,7] Dubrovskyi O,[2,7] Bondarenko G,[2,7] Gaisina I,[8] Yemelyanov A,[3] Procissi D,[5]
James CD,[4] Chandler J,[4] O' Halloran TV,[7] Kozikowski A,[8] Raizer J,[6] Mazar A[1,2,7]

[1]Center for Developmental Therapeutics, [2]Department of Pharmacology, [3]Department of Medicine, [4]Department of Neurological Surgery,
[5]Department of Radiology, [6]Department of Neurology, Feinberg School of Medicine, and [7]Chemistry of Life Processes Institute,
Robert H. Lurie Comprehensive Cancer Center, Northwestern University, Chicago, IL;
[8]College of Pharmacy, Department of Medicinal Chemistry and Pharmacognosy, University of Illinois at Chicago, Chicago, IL

This work was supported in part by P30 CCSG grant (to A.M.), by Brain Tumor Institute grant (to A.M.) and by philantropic support from Baskes Family (to A.M.).

Abstract

Glioblastoma (GBM) is, in essence, an incurable cancer, with most patients surviving 12-15 months following initial diagnosis. Previously published studies identified Glycogen Synthase Kinase-3 (GSK-3) as a new therapeutic target in GBM. Because GSK-3β is a positive regulator of NF-κB-mediated survival and chemoresistance in cancer cells, we hypothesize that the inhibition of GSK-3 may overcome NF-κB-mediated chemoresistance to conventional drugs in human GBM. Using IVIS imaging of live mice, we found that: 1) NF-κB is constitutively active in orthotopic GBM patient-derived xenograft (PDX) tumors expressing an NF-κB luciferase reporter; and 2) a single intravenous injection of our novel GSK-3 inhibitor 9-ING-41 significantly reduced NF-κB transcriptional activity in orthotopic GBM PDX tumor. Using two different GBM PDX tumor models (GBM6 and GBM12), enabled for bioluminescence imaging through luciferase reporter transduction, we evaluated the antitumor effects of 9-ING-41, alone as well as in combination with irinotecan, CCNU and temozolomide. Our *in vivo* results revealed that treatment with a combination of 9-ING-41 and CCNU leads to a complete regression of orthotopic GBM6 and GBM12 tumors. Histological evaluation of mouse brain confirmed the absence of cancer cells in mice treated with 9-ING-41+CCNU. Our results provide a rationale to advance 9-ING-41 for clinical evaluation in treating GBM, especially when combined with CCNU cytotoxic therapy.

Introduction

Malignant brain tumors represent one of the most devastating and incurable cancers. Treatment with conventional chemotherapeutic drugs has had little impact on glioblastoma (GBM) progression. Thus, GBM represents a significant unmet medical need and the identification of new therapeutic agents is urgently needed for the effective treatment of GBM to improve clinical outcomes. The molecular analysis of brain tumor biopsies identified that NF-κB and its target genes are overexpressed in GBM and astrocytoma tumors compared to normal brain tissue (1, 2). In addition, a positive correlation between NF-κB activation and poor GBM prognosis was reported (3). Aberrant NF-κB activity was found critical for GBM invasive phenotype formation and resistance to alkylating agents in GBM (1, 4). Thus, targeting components of NF-κB signaling activation might represent a useful therapeutic approach to overcome malignant brain cancer growth and chemoresistance. Glycogen Synthase Kinase-3β (GSK-3β), a serine/threonine protein kinase, is an essential positive regulator of NF-κB transcriptional activity (5). Our previous studies showed that GSK-3β is a positive regulator of NF-κB-mediated survival and chemoresistance of cancer cells (6-11). Our studies also demonstrated that the inhibition of GSK-3 decreases cancer cell survival via suppression of the NF-κB-mediated expression of Bcl-2 and XIAP (6, 8, 10, 11). Moreover, we have demonstrated that AR-A014418, a toolbox GSK-3 inhibitor, enhanced the anticancer effect of docetaxel and synergistically decreased the viability of renal cancer cells (10). Similarly, AR-A014418 was shown to sensitize pancreatic cancer cells to gemcitabine (12). Finally, a number of published studies identified GSK-3β as a new therapeutic target in human GBM (13-15).

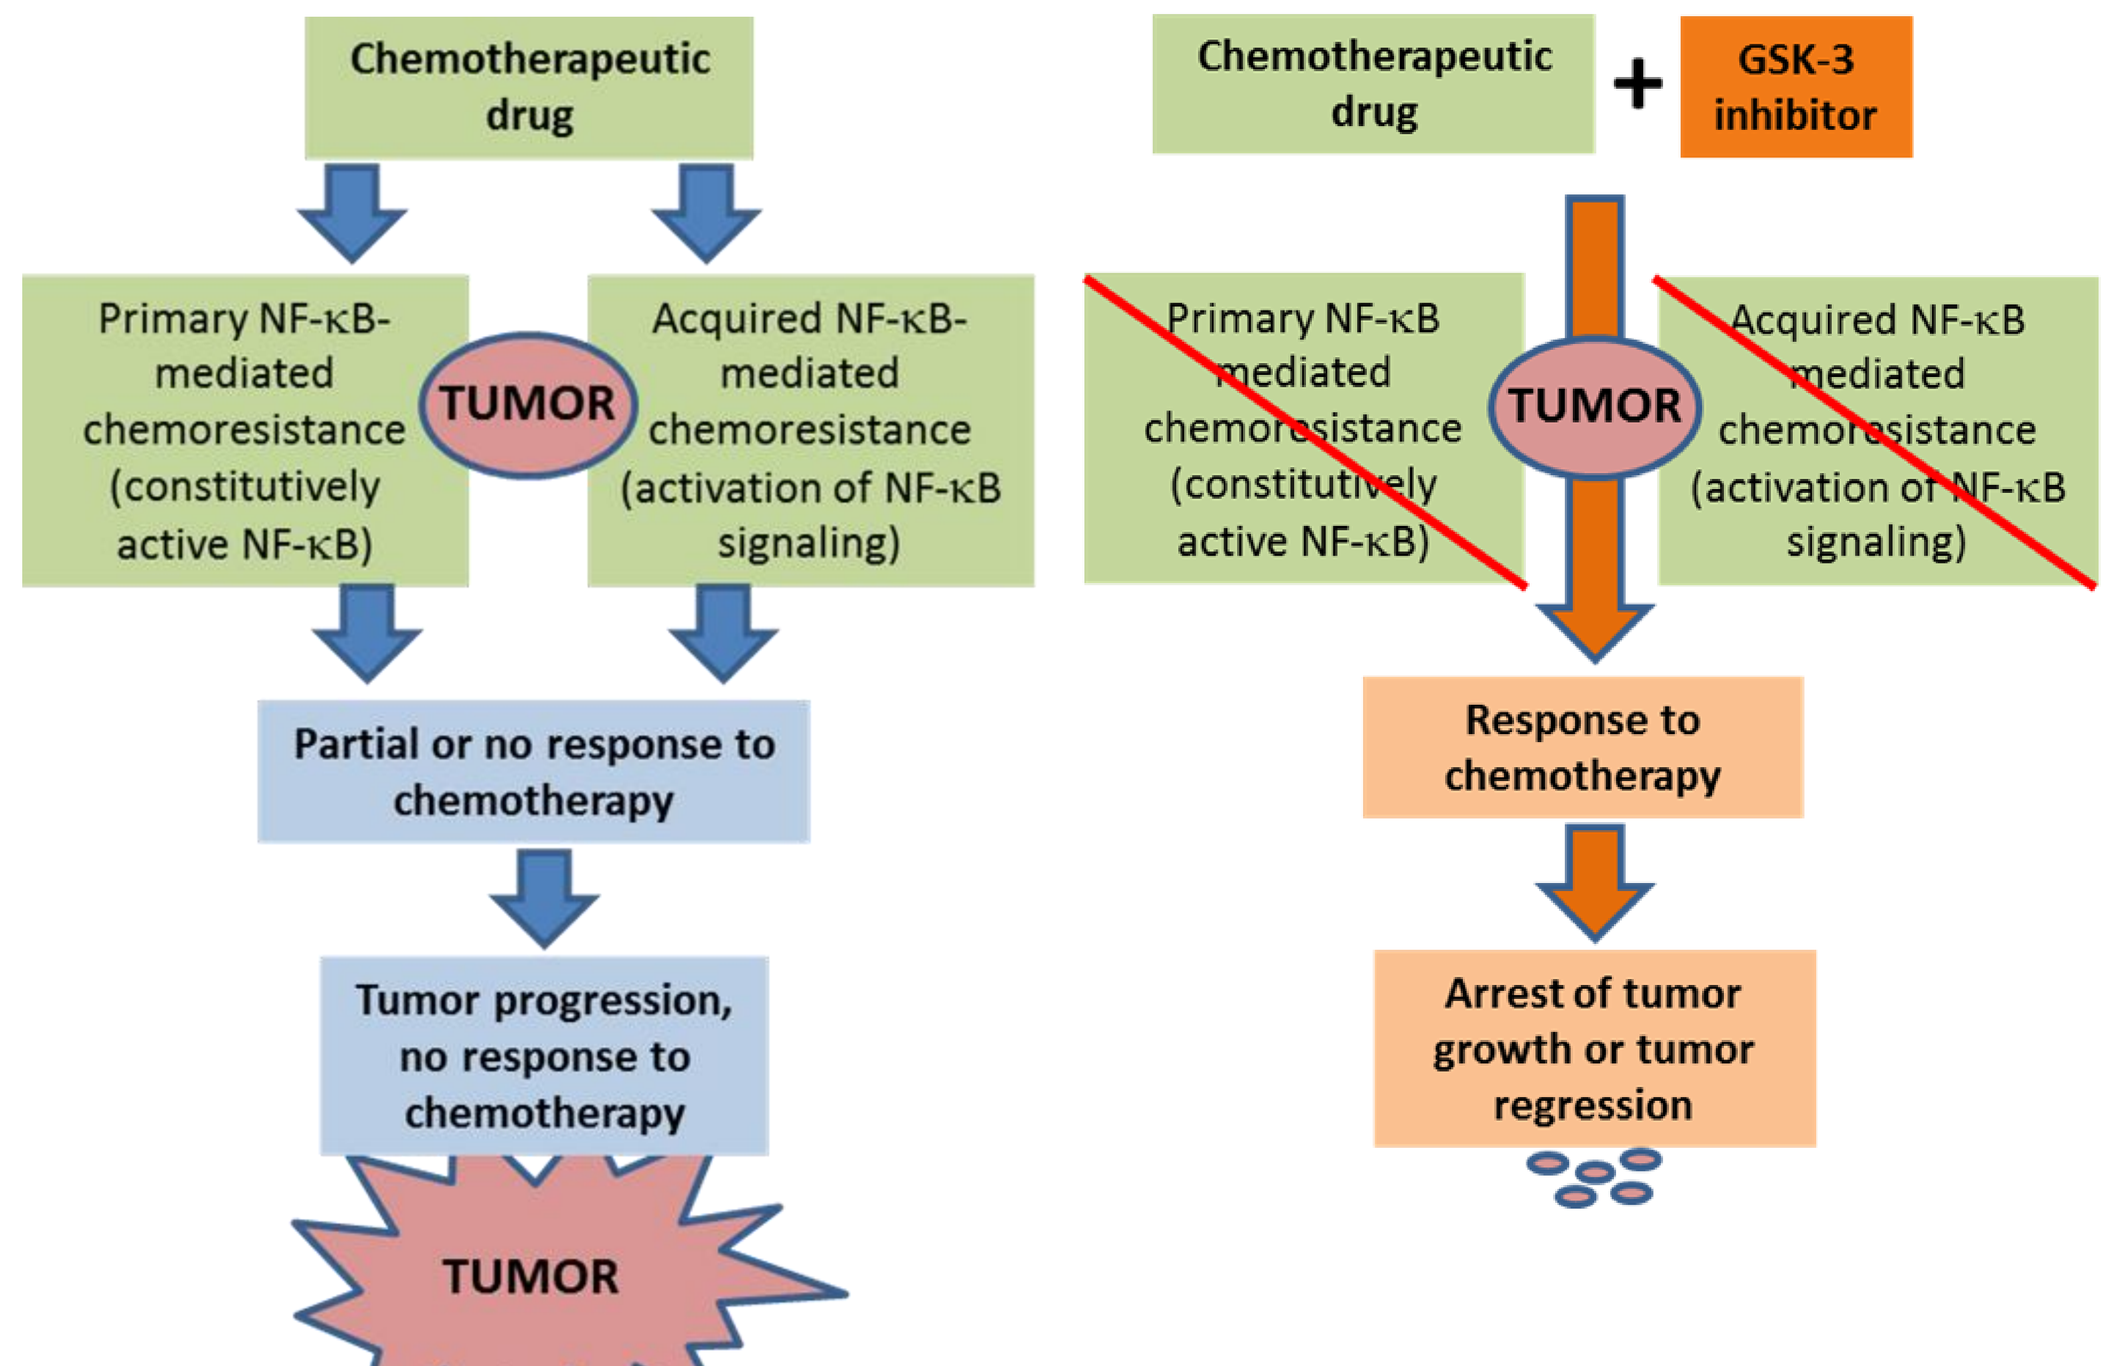

Chemotherapeutic drug
Primary NF-κB-mediated chemoresistance (constitutively active NF-κB)
TUMOR
Acquired NF-κB-mediated chemoresistance (activation of NF-κB signaling)
Partial or no response to chemotherapy
Tumor progression, no response to chemotherapy
TUMOR
Chemotherapeutic drug
+
GSK-3 inhibitor
Primary NF-κB mediated chemoresistance (constitutively active NF-κB)
TUMOR
Acquired NF-κB mediated chemoresistance (activation of NF-κB signaling)
Response to chemotherapy
Arrest of tumor growth or tumor regression

Figure 1. Hypothesis: inhibition of GSK-3β, a positive regulator of NF-κB activity, overcomes NF-κB-mediated chemoresistance and thereby potentiates the effect of conventional chemodrugs in human GBM.

Thus, because GSK-3β is a potential positive regulator of NF-κB-mediated survival and chemoresistance in cancer cells, **we hypothesize that the inhibition of GSK-3 may overcome NF-κB-mediated chemoresistance to conventional chemotherapeutic drugs in human GBM** (Fig. 1).

Results (1)

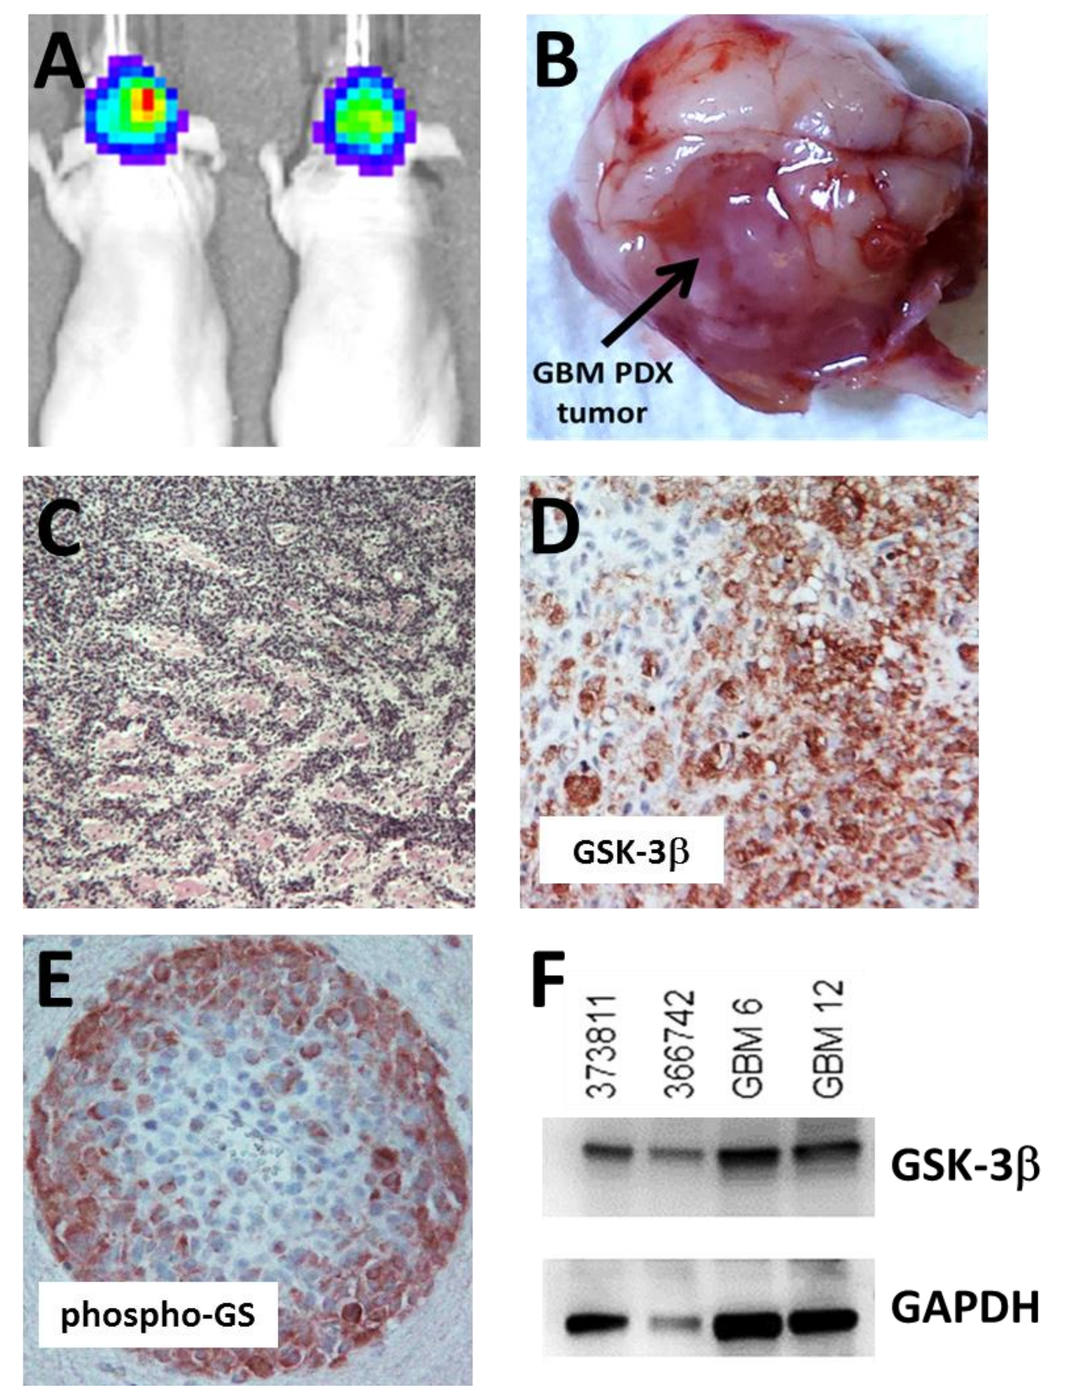

A
B
GBM PDX tumor
C
D
GSK-3β
E
phospho-GS
F
373811
366742
GBM 6
GBM 12
GSK-3β
GAPDH

Figure 2. Development of orthotopic GBM PDX tumor model. *A*, Bioluminescence imaging of mice bearing intracranial tdTomato-Luciferase-transduced GBM PDX tumor (PCF 373811). ***B***, Representative image of GBM PDX tumor (PCF 373811) in resected mouse brain. ***C***, Image of H&E-stained section of orthotopic GBM PDX tumor (PCF 373811). ***D***, Image of GSK-3β expression in orthotopic GBM PDX tumor (PCF 373811). ***E***, Image of tumor immunohistochemical staining for phospho-Glycogen Synthase (GS), a downstream target of GSK-3, in orthotopic GBM PDX tumor (PCF 373811). ***F***, Immunoblot results for subQ GBM PDX tumor expression of GSK-3β.

We developed subcutaneous (subQ) and orthotopic GBM PDX tumor models using freshly resected GBM tumor specimens from brain cancer patients. GBM PDX models preserve the key features and cellular diversity of human GBM tumors. A small piece of GBM PDX tumor was dissociated and the tumor cell suspension was transduced overnight with lentiviral vector encoding fluorescent (tdTomato) and bioluminescent (Luc2) genes *ex vivo*.

Results (2)

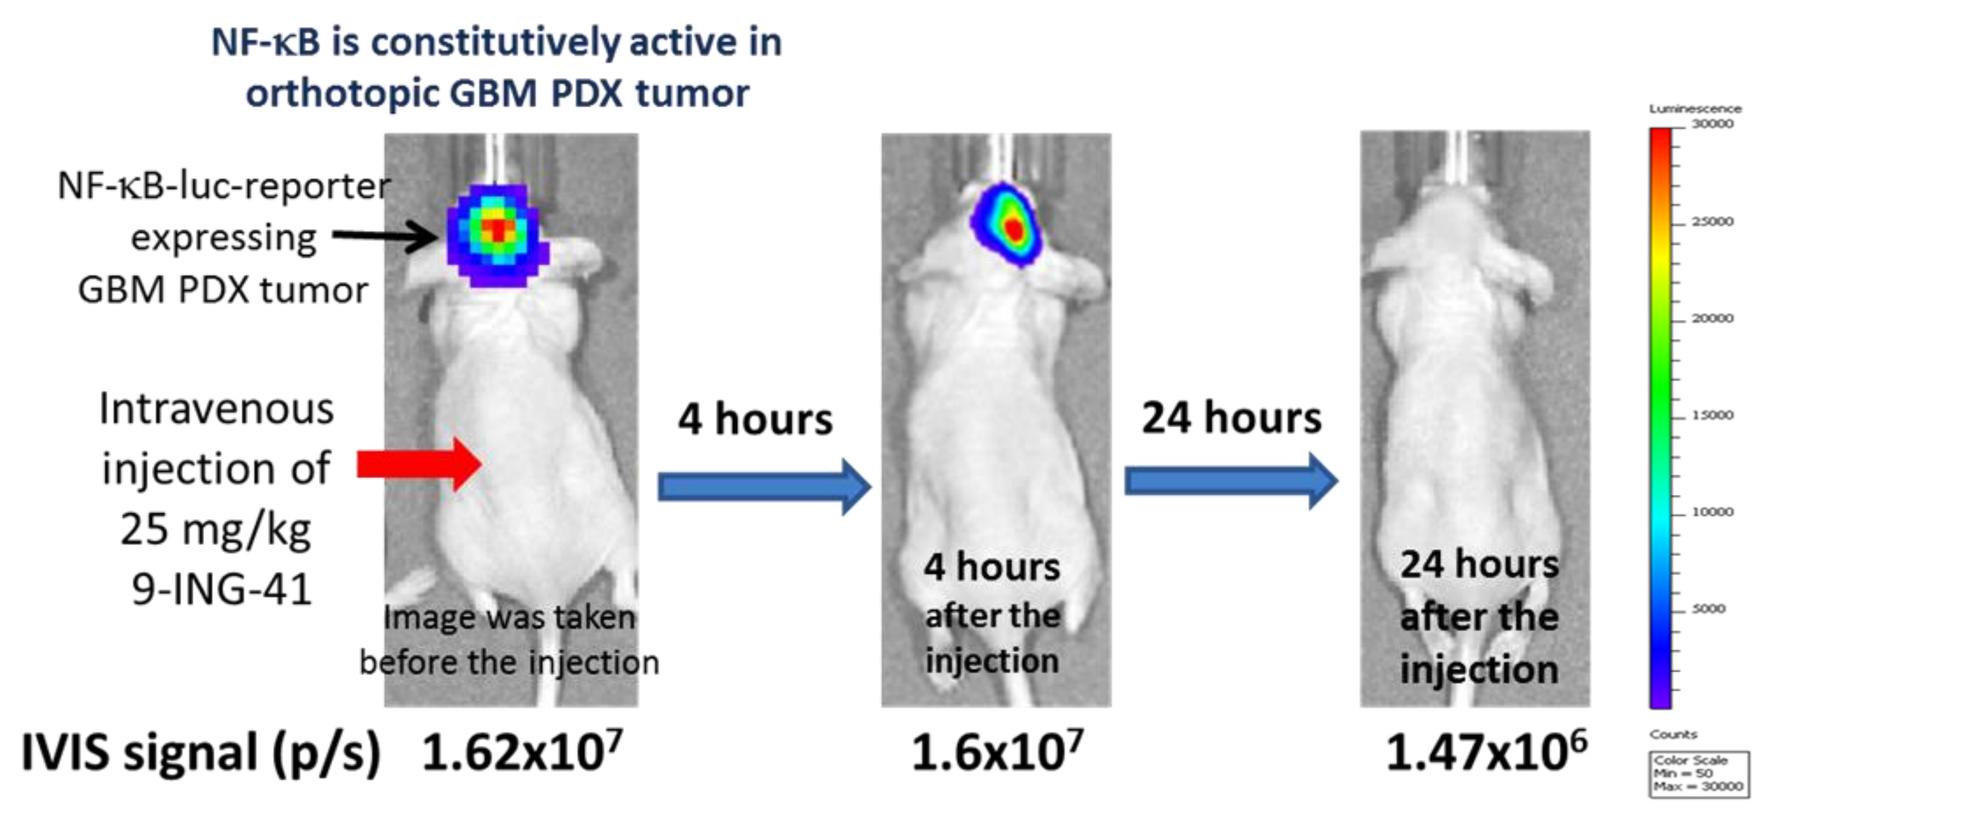

NF-κB is constitutively active in orthotopic GBM PDX tumor
NF-κB-luc-reporter expressing GBM PDX tumor
Intravenous injection of 25 mg/kg 9-ING-41
Image was taken before the injection
4 hours
4 hours after the injection
24 hours
24 hours after the injection
IVIS signal (p/s) 1.62x10^7
1.6x10^7
1.47x10^6

Figure 3. Treatment with GSK-3 inhibitor 9-ING-41 suppresses NF-κB activity in orthotopic GBM PDX tumor (PCF 373811). Bioluminescence imaging of mice bearing intracranial NF-κB-luciferase reporter-transduced GBM PDX tumor (PCF 373811) was performed at indicated time points before and after the treatment with 9-ING-41 as indicated.

We tested whether inhibition of GSK-3β, a positive regulator of NF-κB activity, using our novel and proprietary compound 9-ING-41, overcomes NF-κB-mediated chemoresistance to conventional chemotherapeutic drugs in human GBM. Our goal was to find a combination of 9-ING-41 and cytotoxic chemodrug to achieve the regression of GBM tumor. To determine optimal dosage, schedule and combination of 9-ING-41 with irinotecan, temozolomide and CCNU, we performed a subQ GBM PDX tumor study (Fig. 4). Informed by the results of the subQ experiment, we then tested the most promising combination of 9-ING-41 and CCNU in orthotopic GBM PDX tumor models (Fig. 5-7).

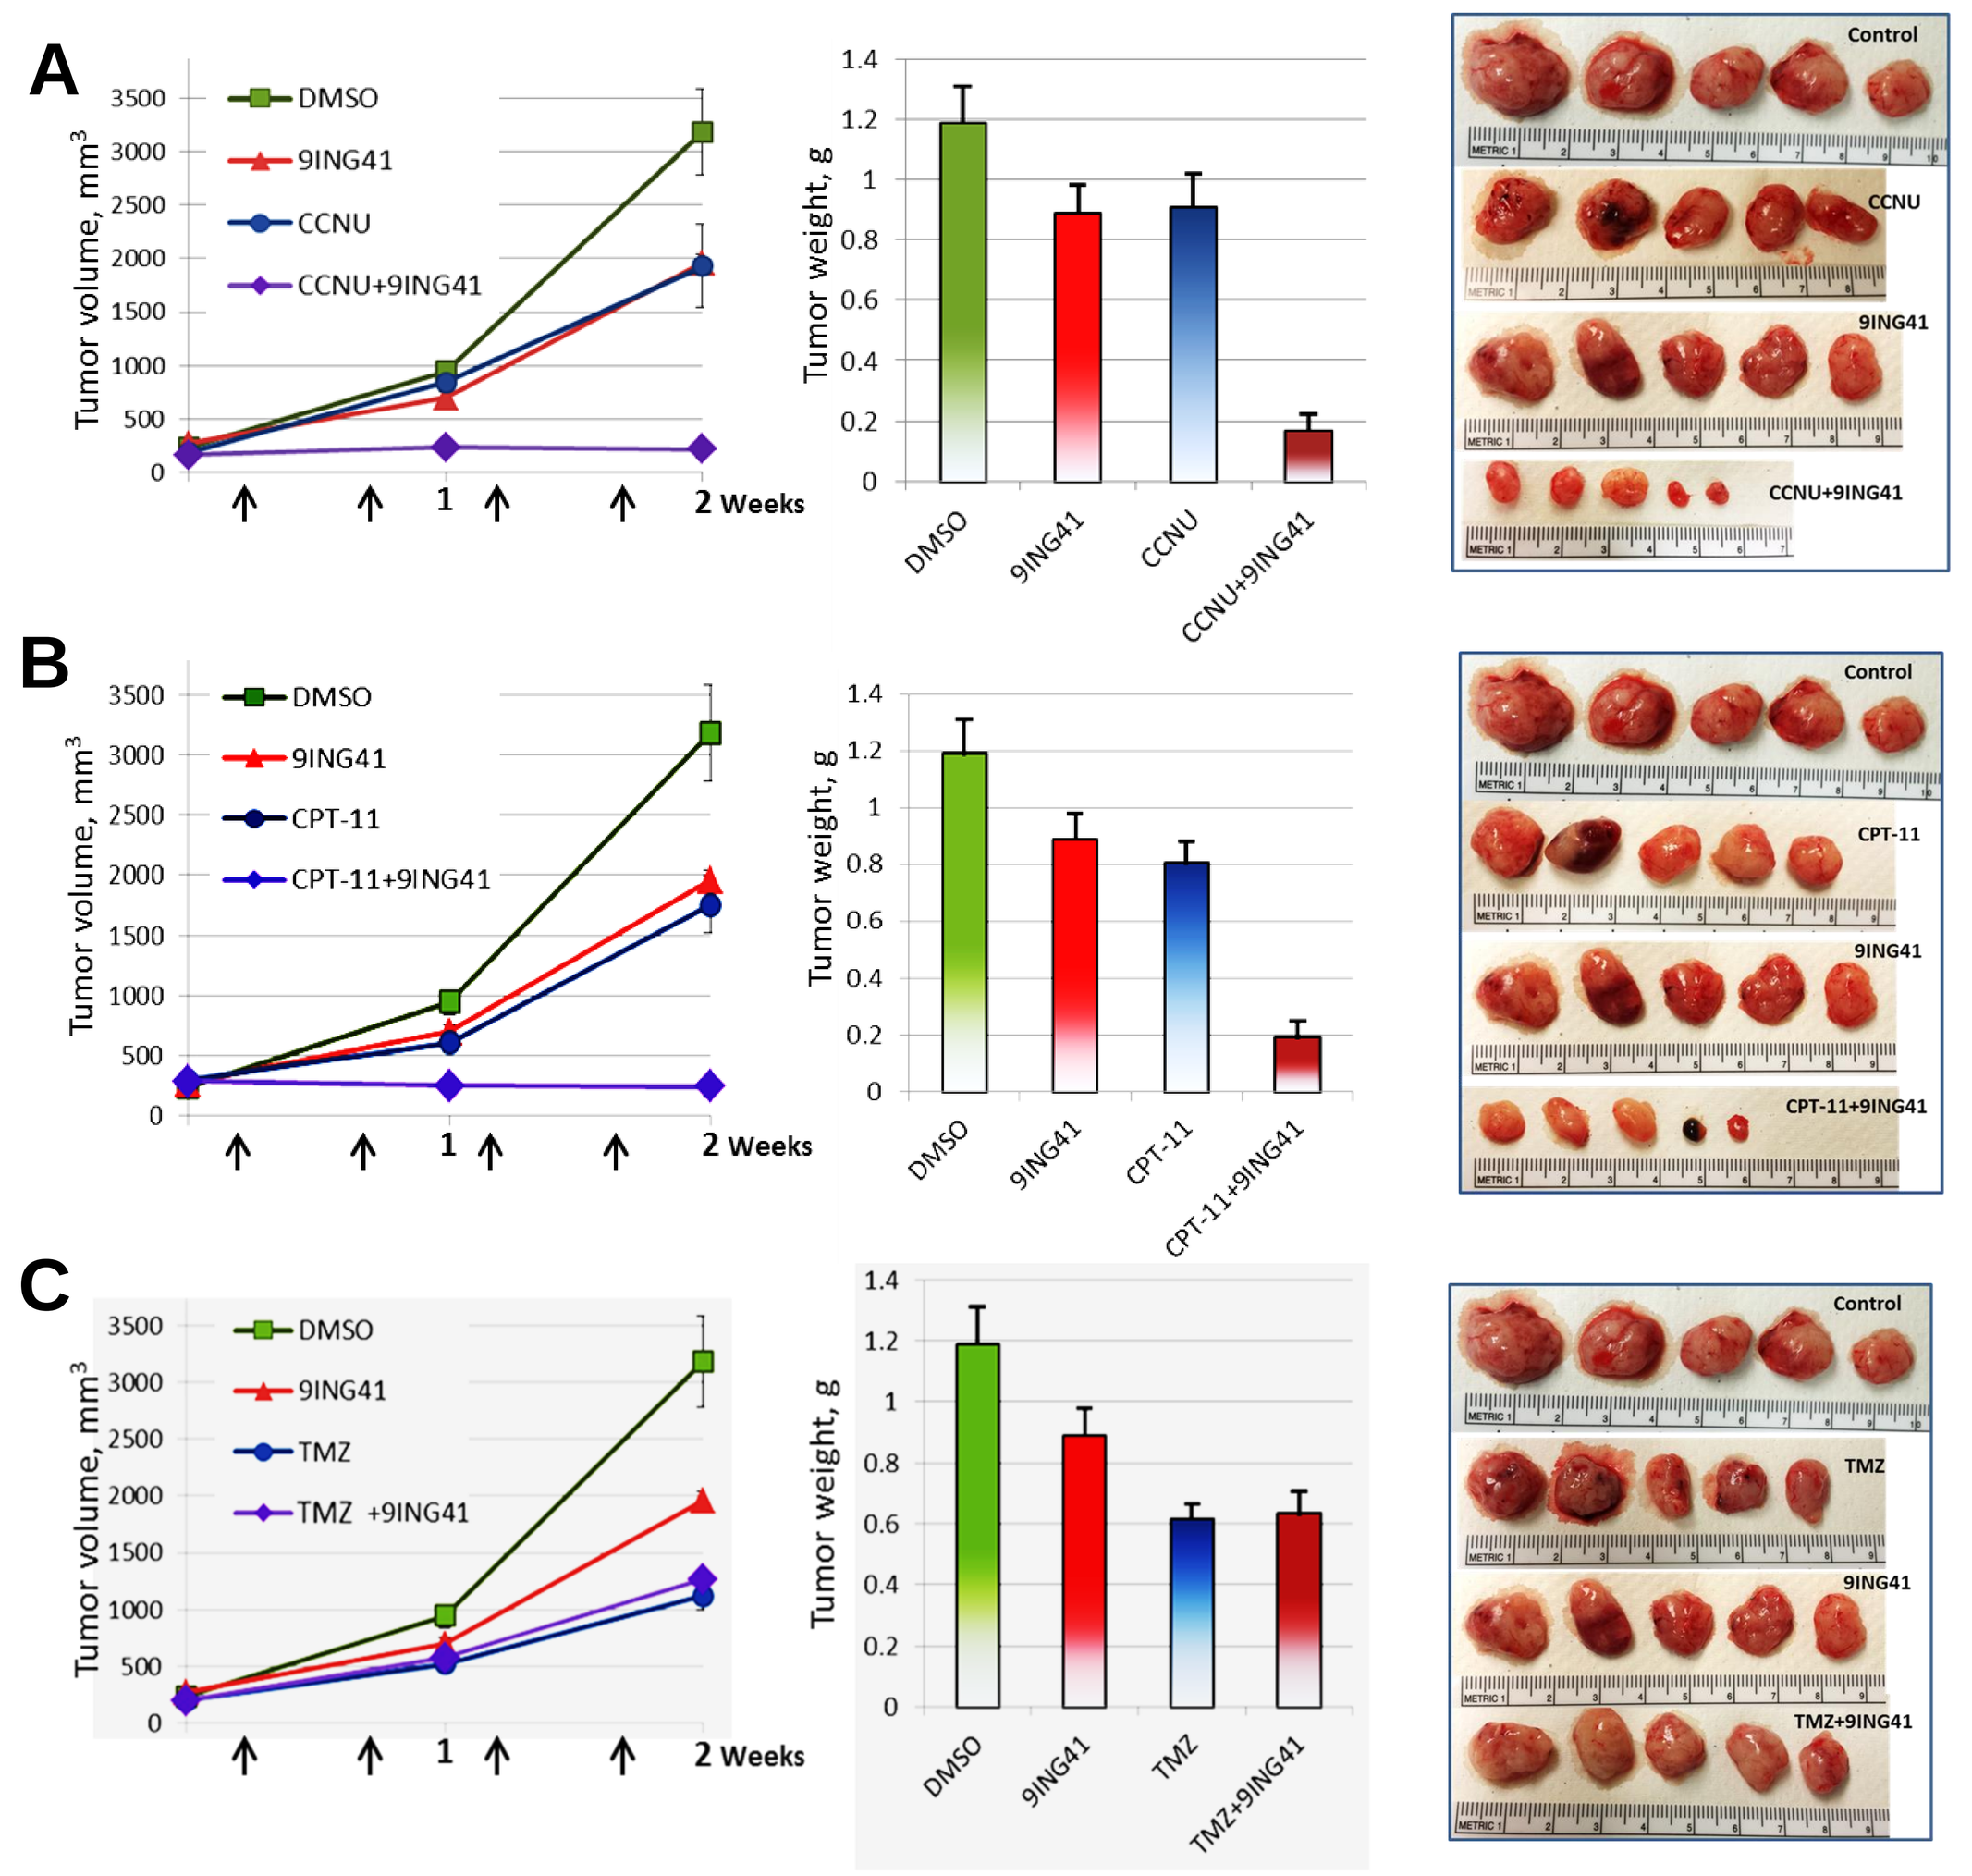

A
DMSO
9ING41
CCNU
CCNU+9ING41
Tumor volume, mm3
Tumor weight, g
2 Weeks
Control
B
CPT-11
CPT-11+9ING41
C
TMZ
TMZ +9ING41
TMZ+9ING41

Figure 4. Treatment with GSK-3 inhibitor 9-ING-41 enhances the antitumor effect of CCNU and CPT-11 in a subQ model of GBM6 PDX tumor. Small pieces of subQ GBM6 PDX tumor were engrafted subQ to nude mice (1 tumor per mouse). Tumors were size matched and mice were randomized into treatment groups (5 mice per group). Vehicle (20 μL DMSO), 70 mg/kg 9ING41, 1 mg/kg CCNU (***A***), 5 mg/kg CPT-11 (***B***) or 1 mg/kg temozolomide (TMZ) (***C***), were injected i.p. at indicated doses as shown by arrows (***A-C***, *left panel*). Mean tumor volumes are plotted; bars, SE. Mice were sacrificed in 2 weeks after initiation of treatment and the weight of resected tumors determined (***A-C***, *middle panel*). Bar graphs: mean tumor weight; SE is indicated. Representative pictures of GBM PDX subQ tumors from each group of animals (***A-C***, *right panel*).

Results (3)

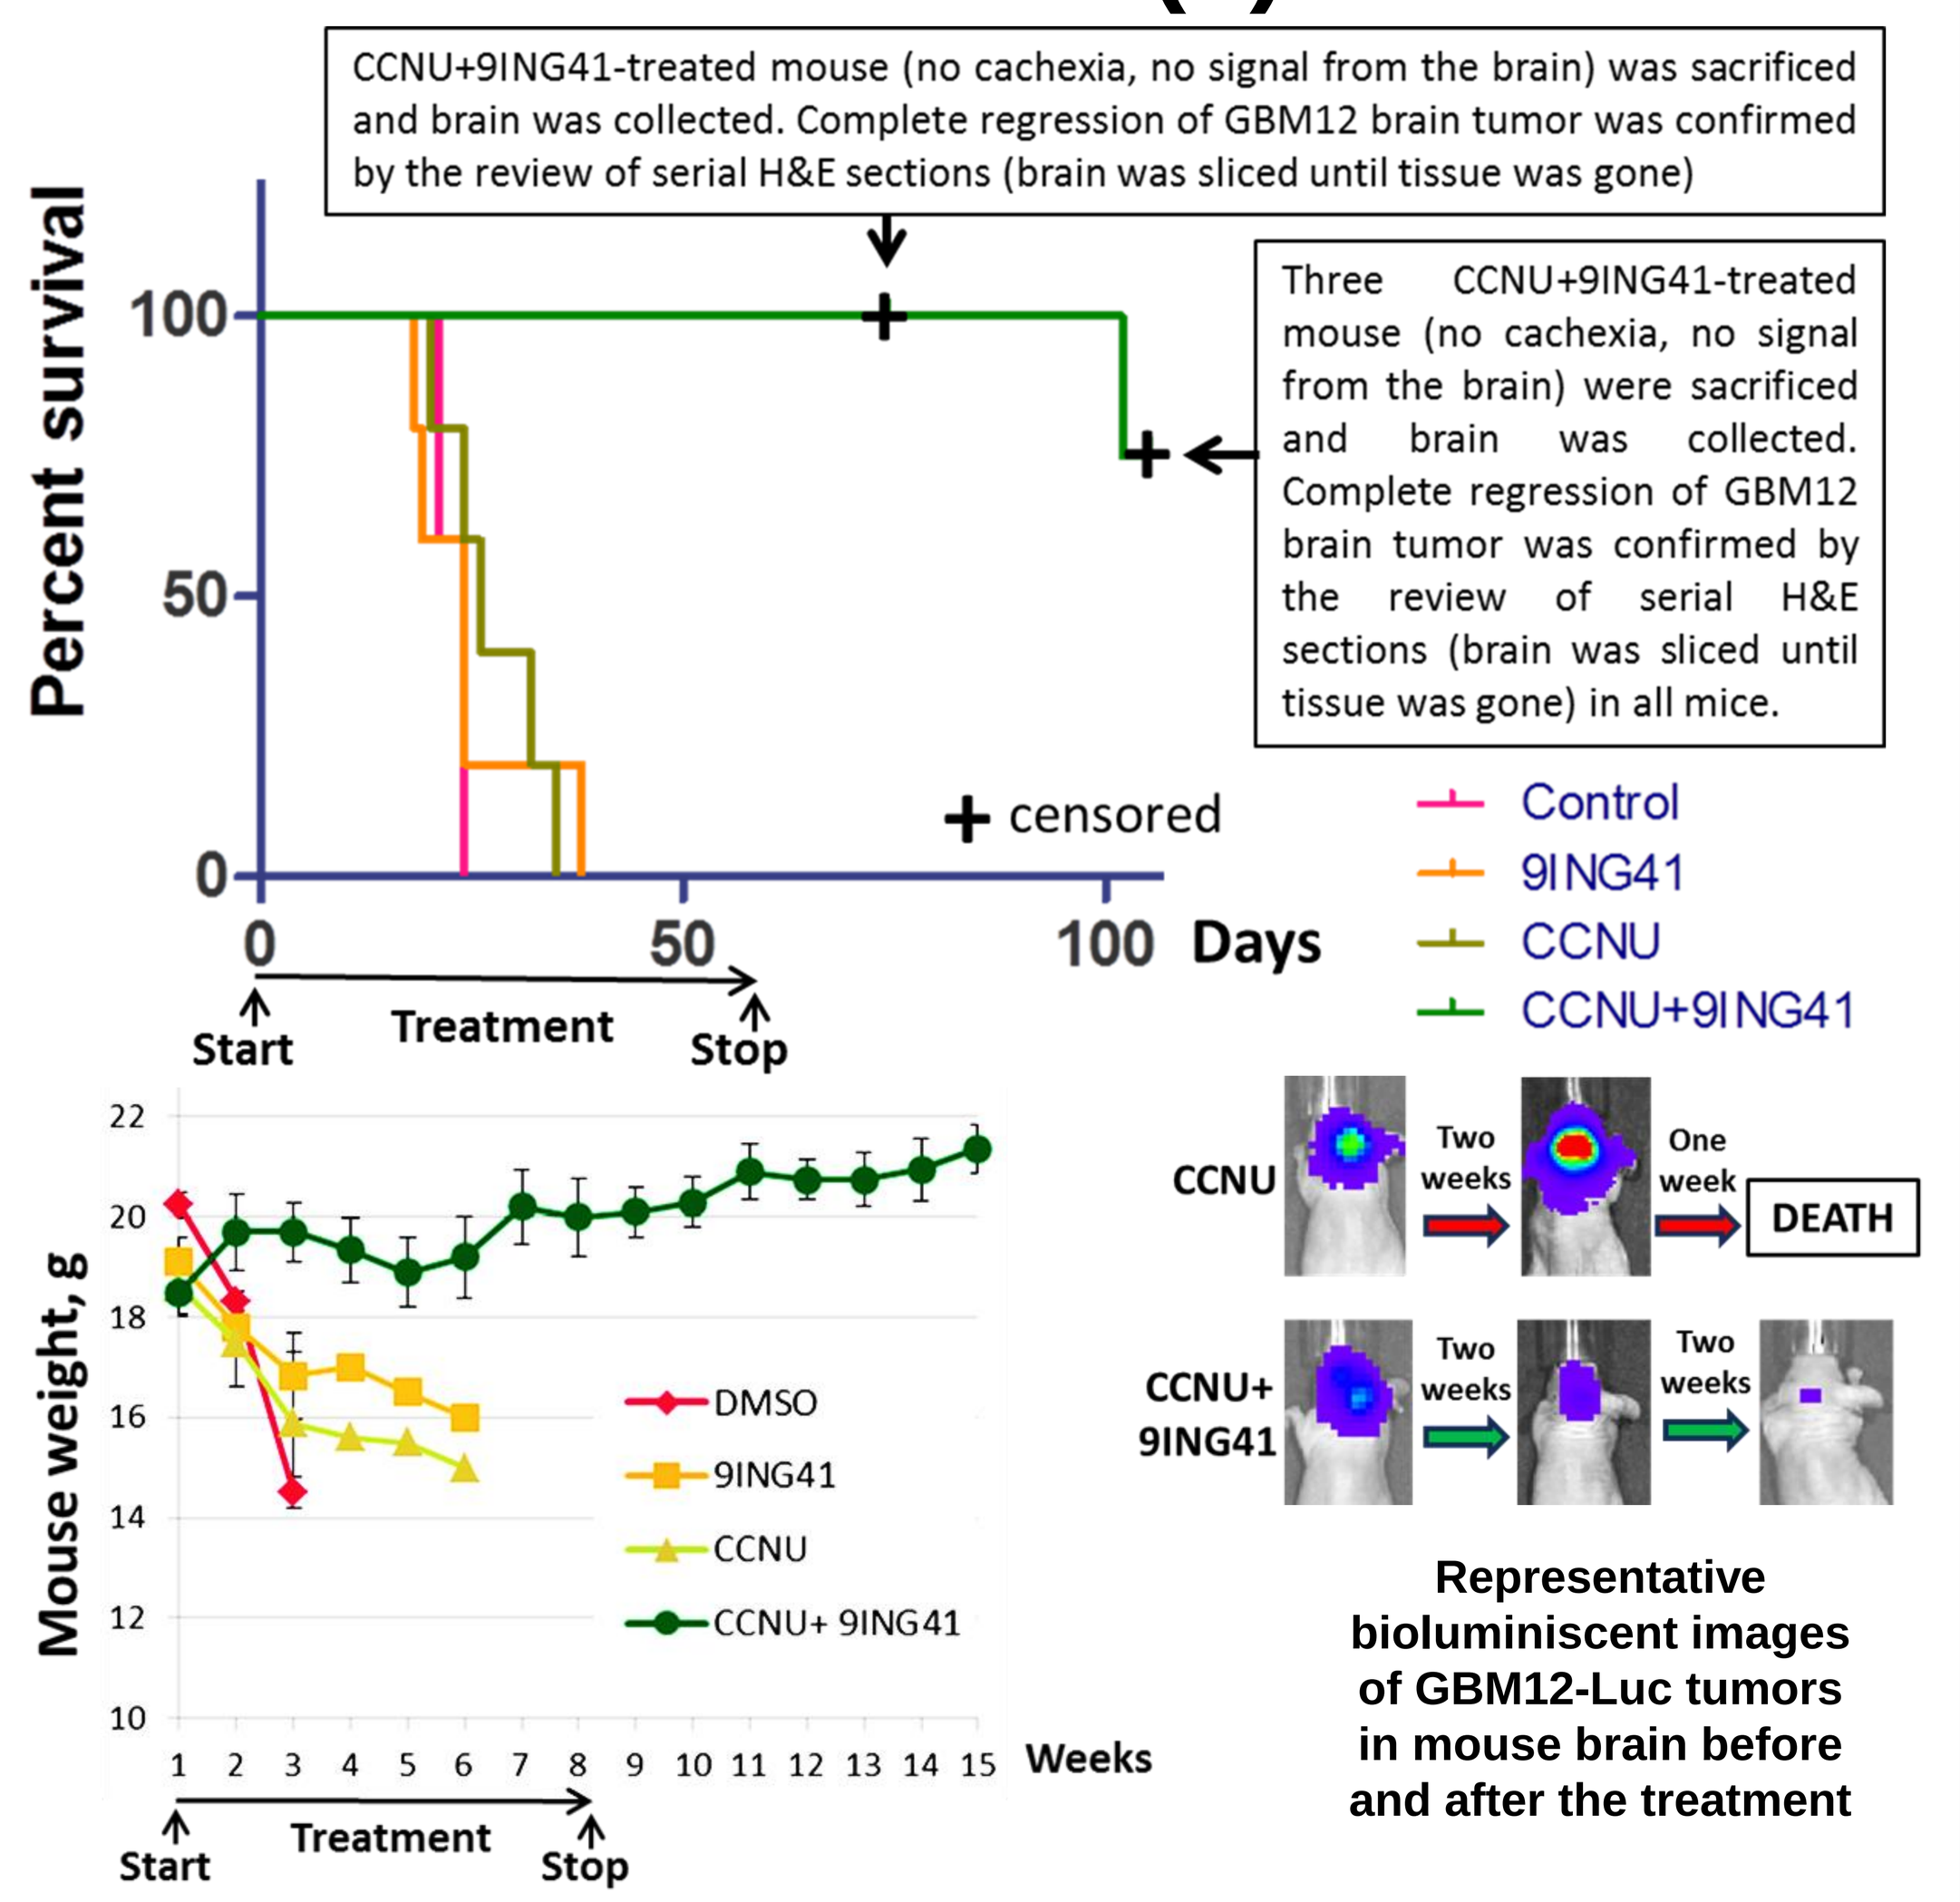

CCNU+9ING41-treated mouse (no cachexia, no signal from the brain) was sacrificed and brain was collected. Complete regression of GBM12 brain tumor was confirmed by the review of serial H&E sections (brain was sliced until tissue was gone)
Three CCNU+9ING41-treated mouse (no cachexia, no signal from the brain) were sacrificed and brain was collected. Complete regression of GBM12 brain tumor was confirmed by the review of serial H&E sections (brain was sliced until tissue was gone) in all mice.
Percent survival
+ censored
Days
Start
Treatment
Stop
Control
9ING41
CCNU
CCNU+9ING41
Mouse weight, g
Weeks
DMSO
9ING41
CCNU
CCNU+ 9ING41
CCNU
Two weeks
One week
DEATH
CCNU+ 9ING41
Two weeks
Representative bioluminiscent images of GBM12-Luc tumors in mouse brain before and after the treatment

Figure 5. Treatment with CCNU+9ING41 causes complete regression of orthotopic GBM12 PDX. *Upper panel,* Kaplan-Meier survival analysis of treated mice bearing intracranial human GBM12 Tom-Luc PDX. Animals were enrolled into a xenotrial and treatment was started when brain tumors were detected (bioluminescent signal) by IVIS imaging. Mice were treated 2 times a week with DMSO (5 animals), 2 mg/kg CCNU (5 animals), 70 mg/kg 9ING41 (5 animals) and CCNU+9ING41 (5 animals). *Lower panel*, all DMSO, CCNU and 9-ING-41 treated mice developed cachexia and became moribund, necessitating animal subject euthanasia. Mean mouse weights are indicated; bars, SE. Combination of CCNU and 9ING41 significantly prolonged survival of animals as compared to CCNU treatment alone. <u>Median survival in the control (DMSO), 9ING41, CCNU and CCNU+9ING41 groups was 23, 25, 27 and 104 days, respectively</u> (Logrank test for trend, P=0.0032).

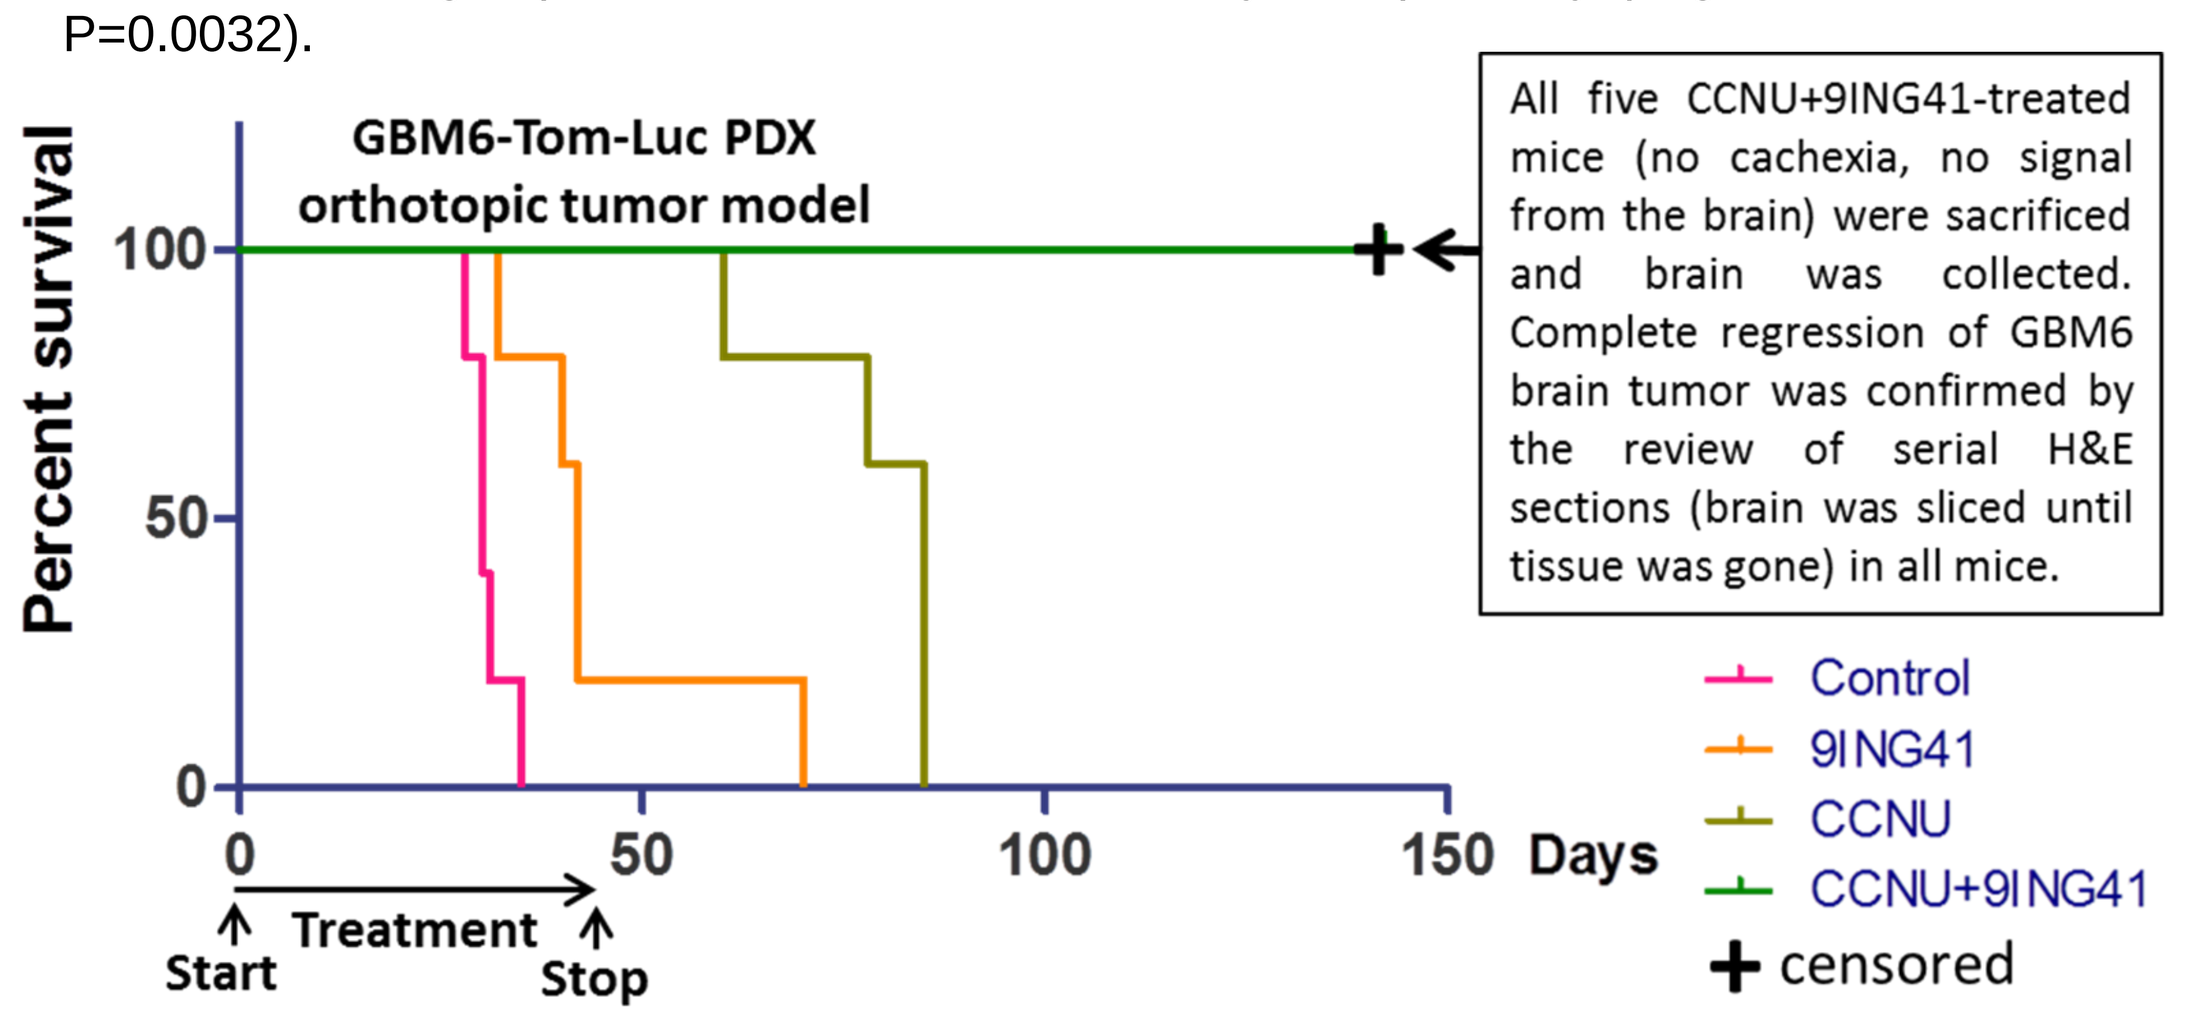

GBM6-Tom-Luc PDX orthotopic tumor model
All five CCNU+9ING41-treated mice (no cachexia, no signal from the brain) were sacrificed and brain was collected. Complete regression of GBM6 brain tumor was confirmed by the review of serial H&E sections (brain was sliced until tissue was gone) in all mice.
Percent survival
Days
Start
Treatment
Stop
Control
9ING41
CCNU
CCNU+9ING41
+ censored

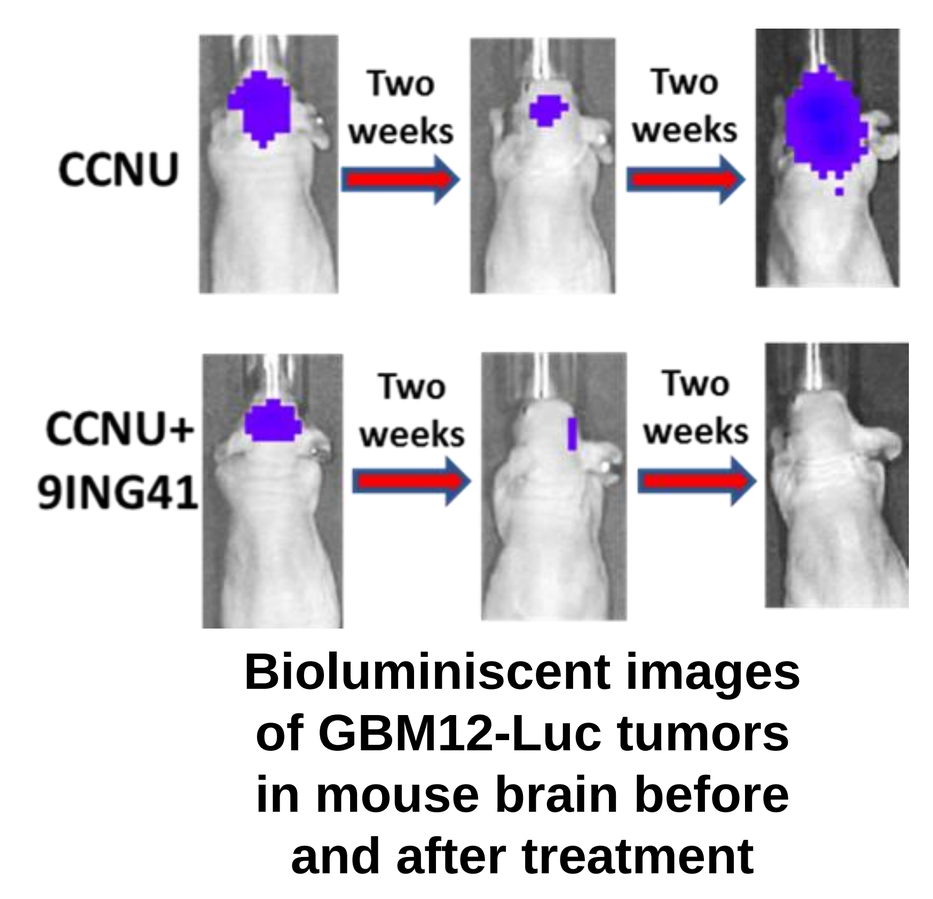

CCNU
Two weeks
CCNU+ 9ING41
Bioluminiscent images of GBM12-Luc tumors in mouse brain before and after treatment

Figure 6. Treatment with CCNU+9ING41 results in complete regression of orthotopic GBM6 PDX tumors. *Upper panel*, Kaplan-Meier survival analysis of treated mice bearing intracranial human GBM6 PDX-Tom-Luc tumors. Animals were enrolled into a xenotrial and treatment was started when brain tumors were detected (bioluminescent signal) by IVIS imaging. Mice were treated 2 times a week with DMSO (5 animals), 2 mg/kg CCNU (5 animals), 70 mg/kg 9ING41 (5 animals) and CCNU+9ING41 (5 animals). Combination of CCNU and 9ING41 significantly prolonged survival of animals as compared to CCNU monotherapy. <u>Median survival in the control (DMSO), 9ING41, CCNU and CCNU+9ING41 groups was 31, 45, 78 and 142 days, respectively</u> (Logrank test for trend, P<0.0001).

Results (4)

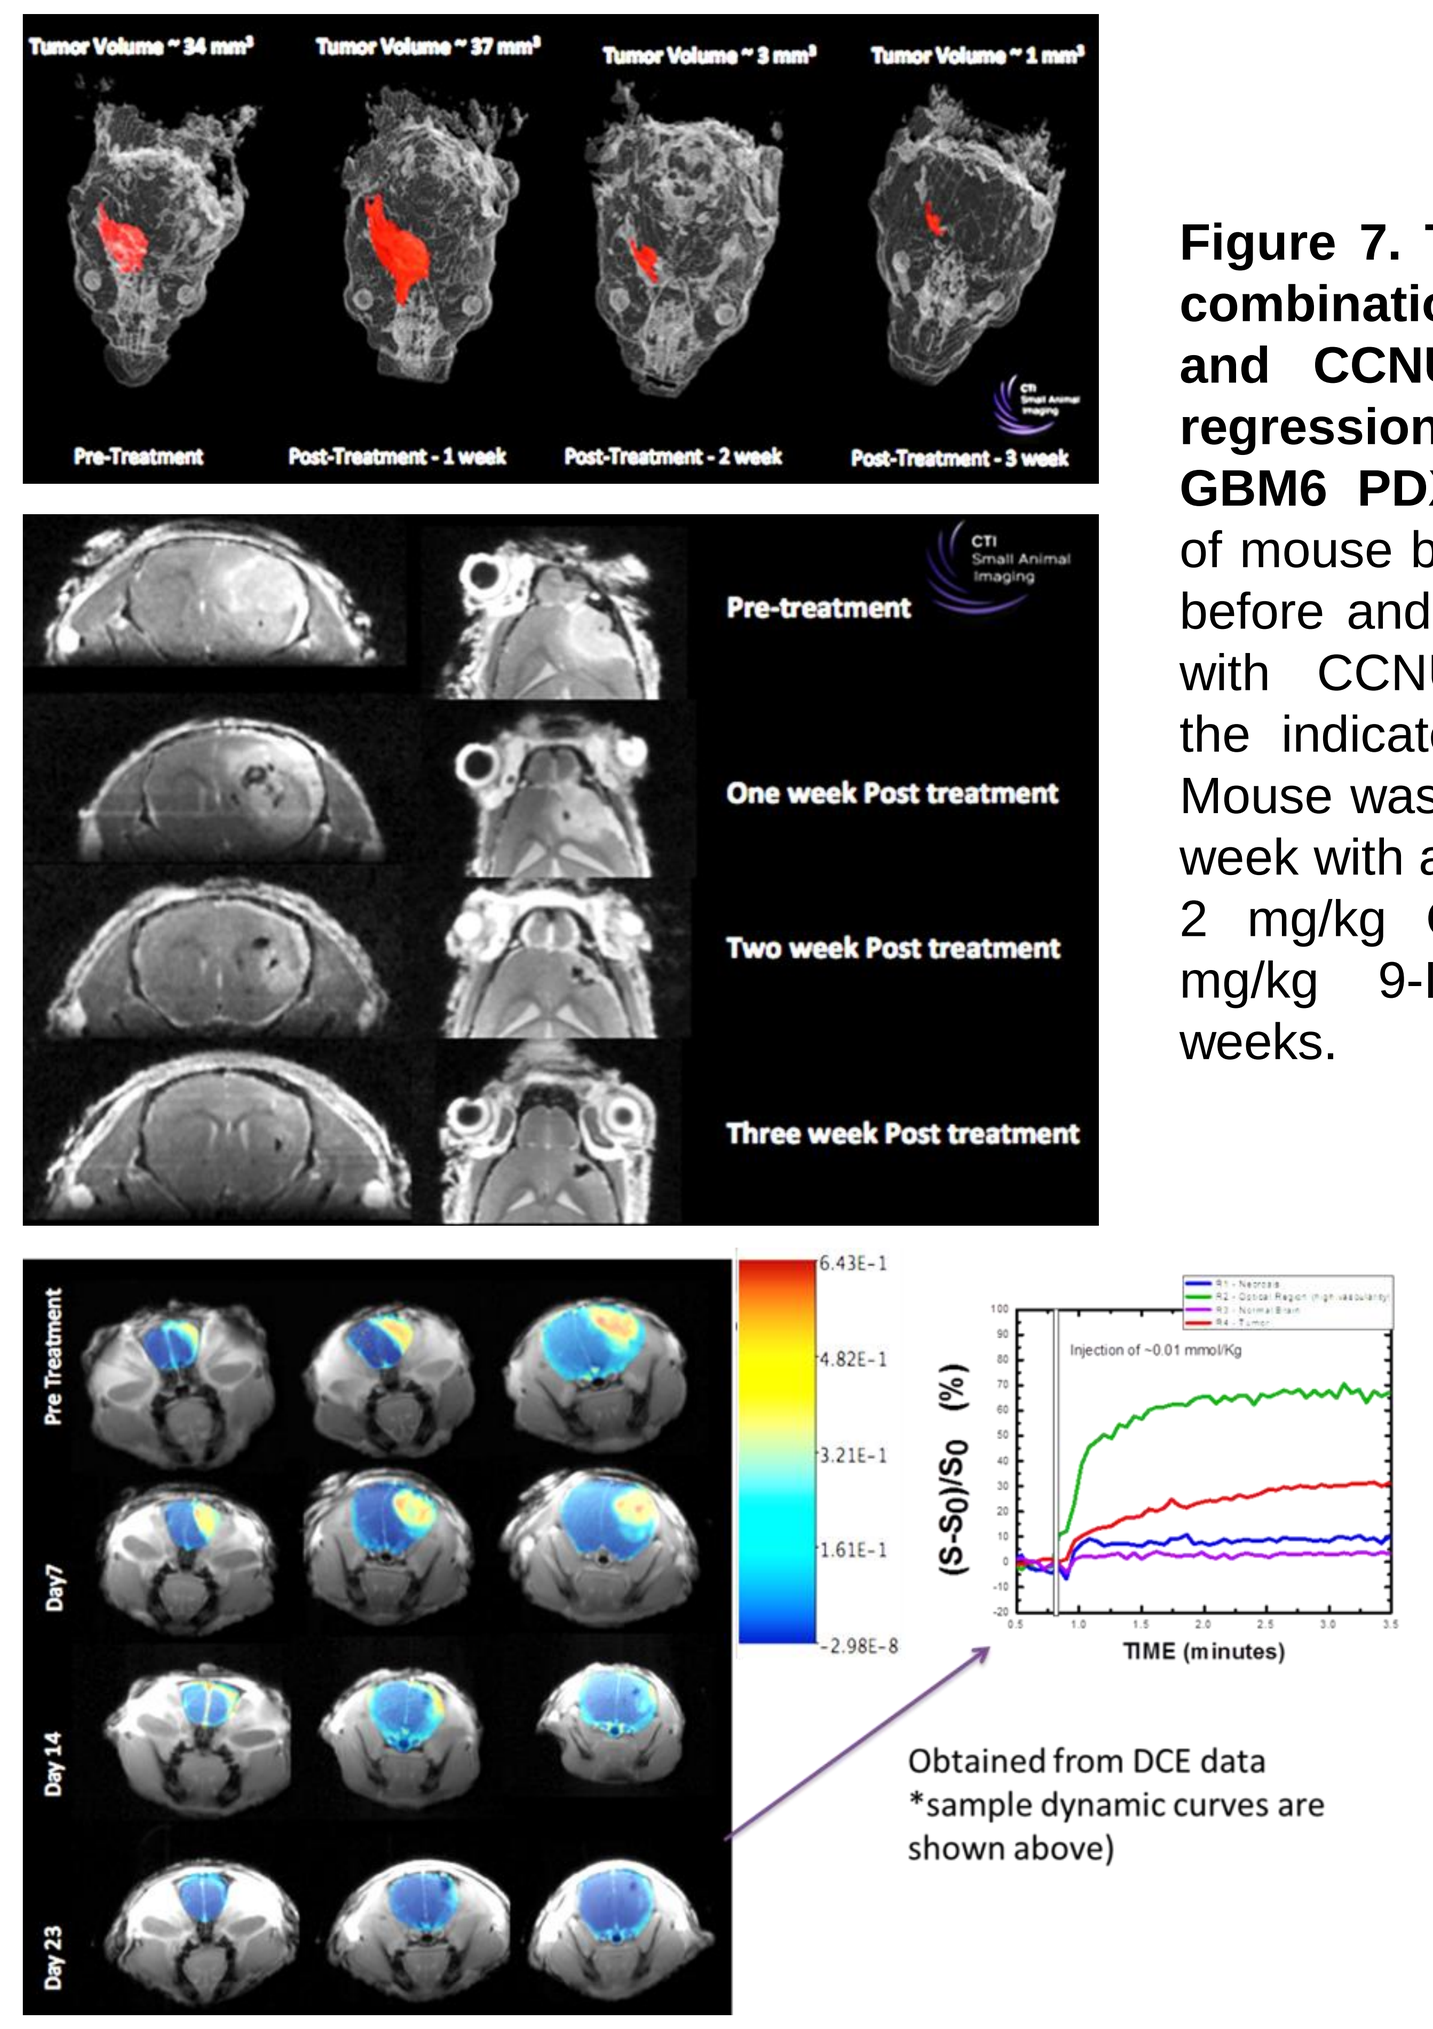

Tumor Volume ~ 34 mm³
Tumor Volume ~ 37 mm³
Tumor Volume ~ 3 mm³
Tumor Volume ~ 1 mm³
Pre-Treatment
Post-Treatment - 1 week
Post-Treatment - 2 week
Post-Treatment - 3 week
Pre-treatment
One week Post treatment
Two week Post treatment
Three week Post treatment
Pre Treatment
Day7
Day 14
Day 23
(S-S0)/S0 (%)
TIME (minutes)
Obtained from DCE data
*sample dynamic curves are shown above)

Figure 7. Treatment with combination of 9-ING-41 and CCNU leads to a regression of intracranial GBM6 PDX. MRI images of mouse brain were taken before and after treatment with CCNU+9-ING-41 at the indicated time points. Mouse was treated twice a week with a combination of 2 mg/kg CCNU and 70 mg/kg 9-ING-41 for 3 weeks.

Conclusions

Clinically, initial GBM response to chemotherapy has two scenarios: 1) refractory tumor with no response, or 2) partial response with subsequent relapse and growth of refractory tumor. Our results demonstrate that GSK-3 inhibitor 9-ING-41 enhances GBM chemosensitivity to CCNU in GBM12 (no response to CCNU) and GBM6 (partial response to CCNU) orthotopic GBM PDX models representing both clinical scenarios. Our results provide a rationale to advance 9-ING-41 for clinical evaluation in combination with CCNU for the treatment of human GBM.

References

1. Raychaudhuri B, Han Y, Lu T, Vogelbaum MA. Aberrant constitutive activation of nuclear factor kappaB in glioblastoma multiforme drives invasive phenotype. J Neurooncol 2007;85:39–47.
2. Robe PA, Bentires-Alj M, Bonif M, Rogister B, Deprez M, Haddada H, et al. In vitro and in vivo activity of the nuclear factor-kappaB inhibitor sulfasalazine in human glioblastomas. Clin Cancer Res 2004;10:5595–5603.
3. Brown RE, Law A. Morphoproteomic demonstration of constitutive nuclear factor-kappaB activation in glioblastoma multiforme with genomic correlates and therapeutic implications. Ann Clin Lab Sci 2006;36:421–426.
4. Bredel M, Bredel C, Juric D, Duran GE, Yu RX, Harsh GR, et al. Tumor necrosis factor-alpha-induced protein 3 as a putative regulator of nuclear factor-kappaB-mediated resistance to O6-alkylating agents in human glioblastomas. J Clin Oncol 2006;24:274–287.
5. Hoeflich K, Luo J, Rubie E, Tsao M, Jin O, Woodgett J. Requirement for glycogen synthase kinase-3β in cell survival and NF-κB activation. Nature 2000;406:86–90.
6. Ougolkov A, Fernandez-Zapico M, Savoy D, Urrutia R, Billadeau D. Glycogen synthase kinase-3beta participates in nuclear factor kappaB-mediated gene transcription and cell survival in pancreatic cancer cells. Cancer Res 2005;65:2076-2081.
7. Ougolkov A, Fernandez-Zapico M, Bilim V, Smyrk T, Chari S, Billadeau D. Aberrant nuclear accumulation of glycogen synthase kinase-3beta in human pancreatic cancer: association with kinase activity and tumor dedifferentiation. Clin Cancer Res 2006;12:5074-5081.
8. Ougolkov A, Bone N, Fernandez-Zapico M, Kay N, Billadeau D. Inhibition of glycogen synthase kinase-3 activity leads to epigenetic silencing of nuclear factor kappaB target genes and induction of apoptosis in chronic lymphocytic leukemia B cells. Blood 2007;110:735-742.
9. Ougolkov A, Shakoori A, Zhang B, Modarressi M, Billadeau D, Mai M, et al. Deregulated GSK3beta activity in colorectal cancer: its association with tumor cell survival and proliferation. Biochem Biophys Res Commun 2005;334:1365-1373.
10. Bilim V, Ougolkov A, Yuuki K, Naito S, Kawazoe H, Muto A, et al. Glycogen synthase kinase-3: a new therapeutic target in renal cell carcinoma. Br J Cancer 2009;101:2005-2014.
11. Naito S, Bilim V, Yuuki K, Ugolkov A, Motoyama T, Nagaoka A, et al. Glycogen synthase kinase-3beta: a prognostic marker and a potential therapeutic target in human bladder cancer. Clin Cancer Res 2010;16:5124-5132.
12. Shimasaki T, Ishigaki Y, Nakamura Y, Takata T, Nakaya N, Nakajima H, et al. Glycogen synthase kinase- 3β inhibition sensitizes pancreatic cancer cells to gemcitabine. J Gastroenterol. 2012;47:321-333.
13. Kotliarova S, Pastorino S, Kovell L, Kotliarov Y, Song H, Zhang W, et al. Glycogen synthase kinase-3 inhibition induces glioma cell death through c-MYC, NF-kappaB, and glucose regulation. Cancer Res 2008;68:6643-6651.
14. Miyashita K, Kawakami K, Nakada M, Mai W, Shakoori A, Fujisawa H, et al. Potential therapeutic effect of glycogen synthase kinase 3beta inhibition against human glioblastoma. Clin Cancer Res 2009;15:887-897.
15. Korur S, Huber RM, Sivasankaran B, Petrich M, Morin P Jr, Hemmings BA, et al. GSK3beta regulates differentiation and growth arrest in glioblastoma. PLoS One 2009;4:e7443.