Targeting GSK-3: a new approach for the treatment of neuroblastoma

Dubrovskyi O,[1,3] Ugolkov A,[2,3] Gaisina I,[5] Bondarenko G,[1,3] Strizzi L,[4] Margaryan NV,[4] Kozikowski A,[5] O'Halloran TV,[3] Hendrix MJC,[4] Mazar AM[1,2,3]

[1]Department of Pharmacology, Feinberg School of Medicine, [2]Center for Developmental Therapeutics,
[3]Chemistry for Life Processes Institute, and [4]Stanley Manne Children's Research Institute,
Robert H. Lurie Comprehensive Cancer Center, Northwestern University, Chicago, IL;
[5]College of Pharmacy, Department of Medicinal Chemistry and Pharmacognosy, University of Illinois at Chicago, Chicago, IL

This work was supported in part by P30 CCSG grant (to A.M.) and philantropic support from Little Heroes Children's Cancer Research Fund (to M.H. and A.M.).

Abstract

Neuroblastoma is a devastating pediatric cancer and most patients older than 18 months present with multi-organ metastatic disease. High grade or recurrent disease is refractory to treatment with chemotherapy and almost uniformly fatal. Glycogen Synthase Kinase-3 (GSK-3) was recently identified as a potential therapeutic target in human neuroblastoma. Because GSK-3β has been shown to be a positive regulator of NF-κB-mediated survival and chemoresistance in cancer cells, we hypothesize that the inhibition of GSK-3 may have therapeutic effects in neuroblastoma. Using chemically distinct GSK-3 inhibitors (AR-A014418, TDZD8 and 9-ING-41), we found that pharmacological inhibition of GSK-3 led to a decrease in viability of neuroblastoma cells. However, our novel and proprietary GSK-3 inhibitor 9-ING-41 was identified as being most potent. We observed that inhibition of GSK-3 results in decreased expression of the antiapoptotic molecule XIAP (NF-κB target gene) and a subsequent increase in neuroblastoma cell apoptosis. Our xenograft in vivo studies show that the combination of irinotecan (CPT-11) and 9-ING-41 leads to regression of subcutaneous SK-N-DZ neuroblastoma xenograft tumors at doses of irinotecan that are below the maximum tolerated dose whereas irinotecan or 9-ING-41 monotherapy had little or only modest effects on tumor growth. Our results suggest that the inhibition of GSK-3 is a promising new approach for the treatment of neuroblastoma, especially when combined with irinotecan cytotoxic therapy

Introduction

Neuroblastoma is a devastating pediatric cancer and most patients older than 18 months present with multi-organ metastatic disease. High grade or recurrent disease is refractory to treatment with chemotherapy and almost uniformly fatal. Thus, neuroblastoma represents a significant unmet medical need and the identification of new therapeutic agents is urgently needed for the effective treatment of neuroblastoma to improve clinical outcomes.

NF-κB activation is known to promote human cancer progression, metastasis, and chemoresistance (1, 2). Glycogen Synthase Kinase-3β (GSK-3β), a serine/threonine protein kinase, is an essential positive regulator of NF-κB transcriptional activity (3). Our previous studies showed that GSK-3β is a positive regulator of NF-κB-mediated survival and chemoresistance of cancer cells (4-9). It has been shown that treatment of neuroblastoma cells with doxorubicin or etoposide resulted in enhanced NF-κB transcriptional activity in a dose-dependent manner (10). Our previous studies have demonstrated that the inhibition of GSK-3 decreases cancer cell survival via suppression of the NF-κB-mediated expression of Bcl-2 and XIAP (4-9). Moreover, we have demonstrated that AR-A014418, a toolbox GSK-3 inhibitor, enhanced the anticancer effect of docetaxel and synergistically decreased the viability of renal cancer cells (8). Similarly, AR-A014418 was shown to sensitize pancreatic cancer cells to gemcitabine (11). Finally, a number of published studies identified GSK-3β as a new therapeutic target in human neuroblastoma (12-13).

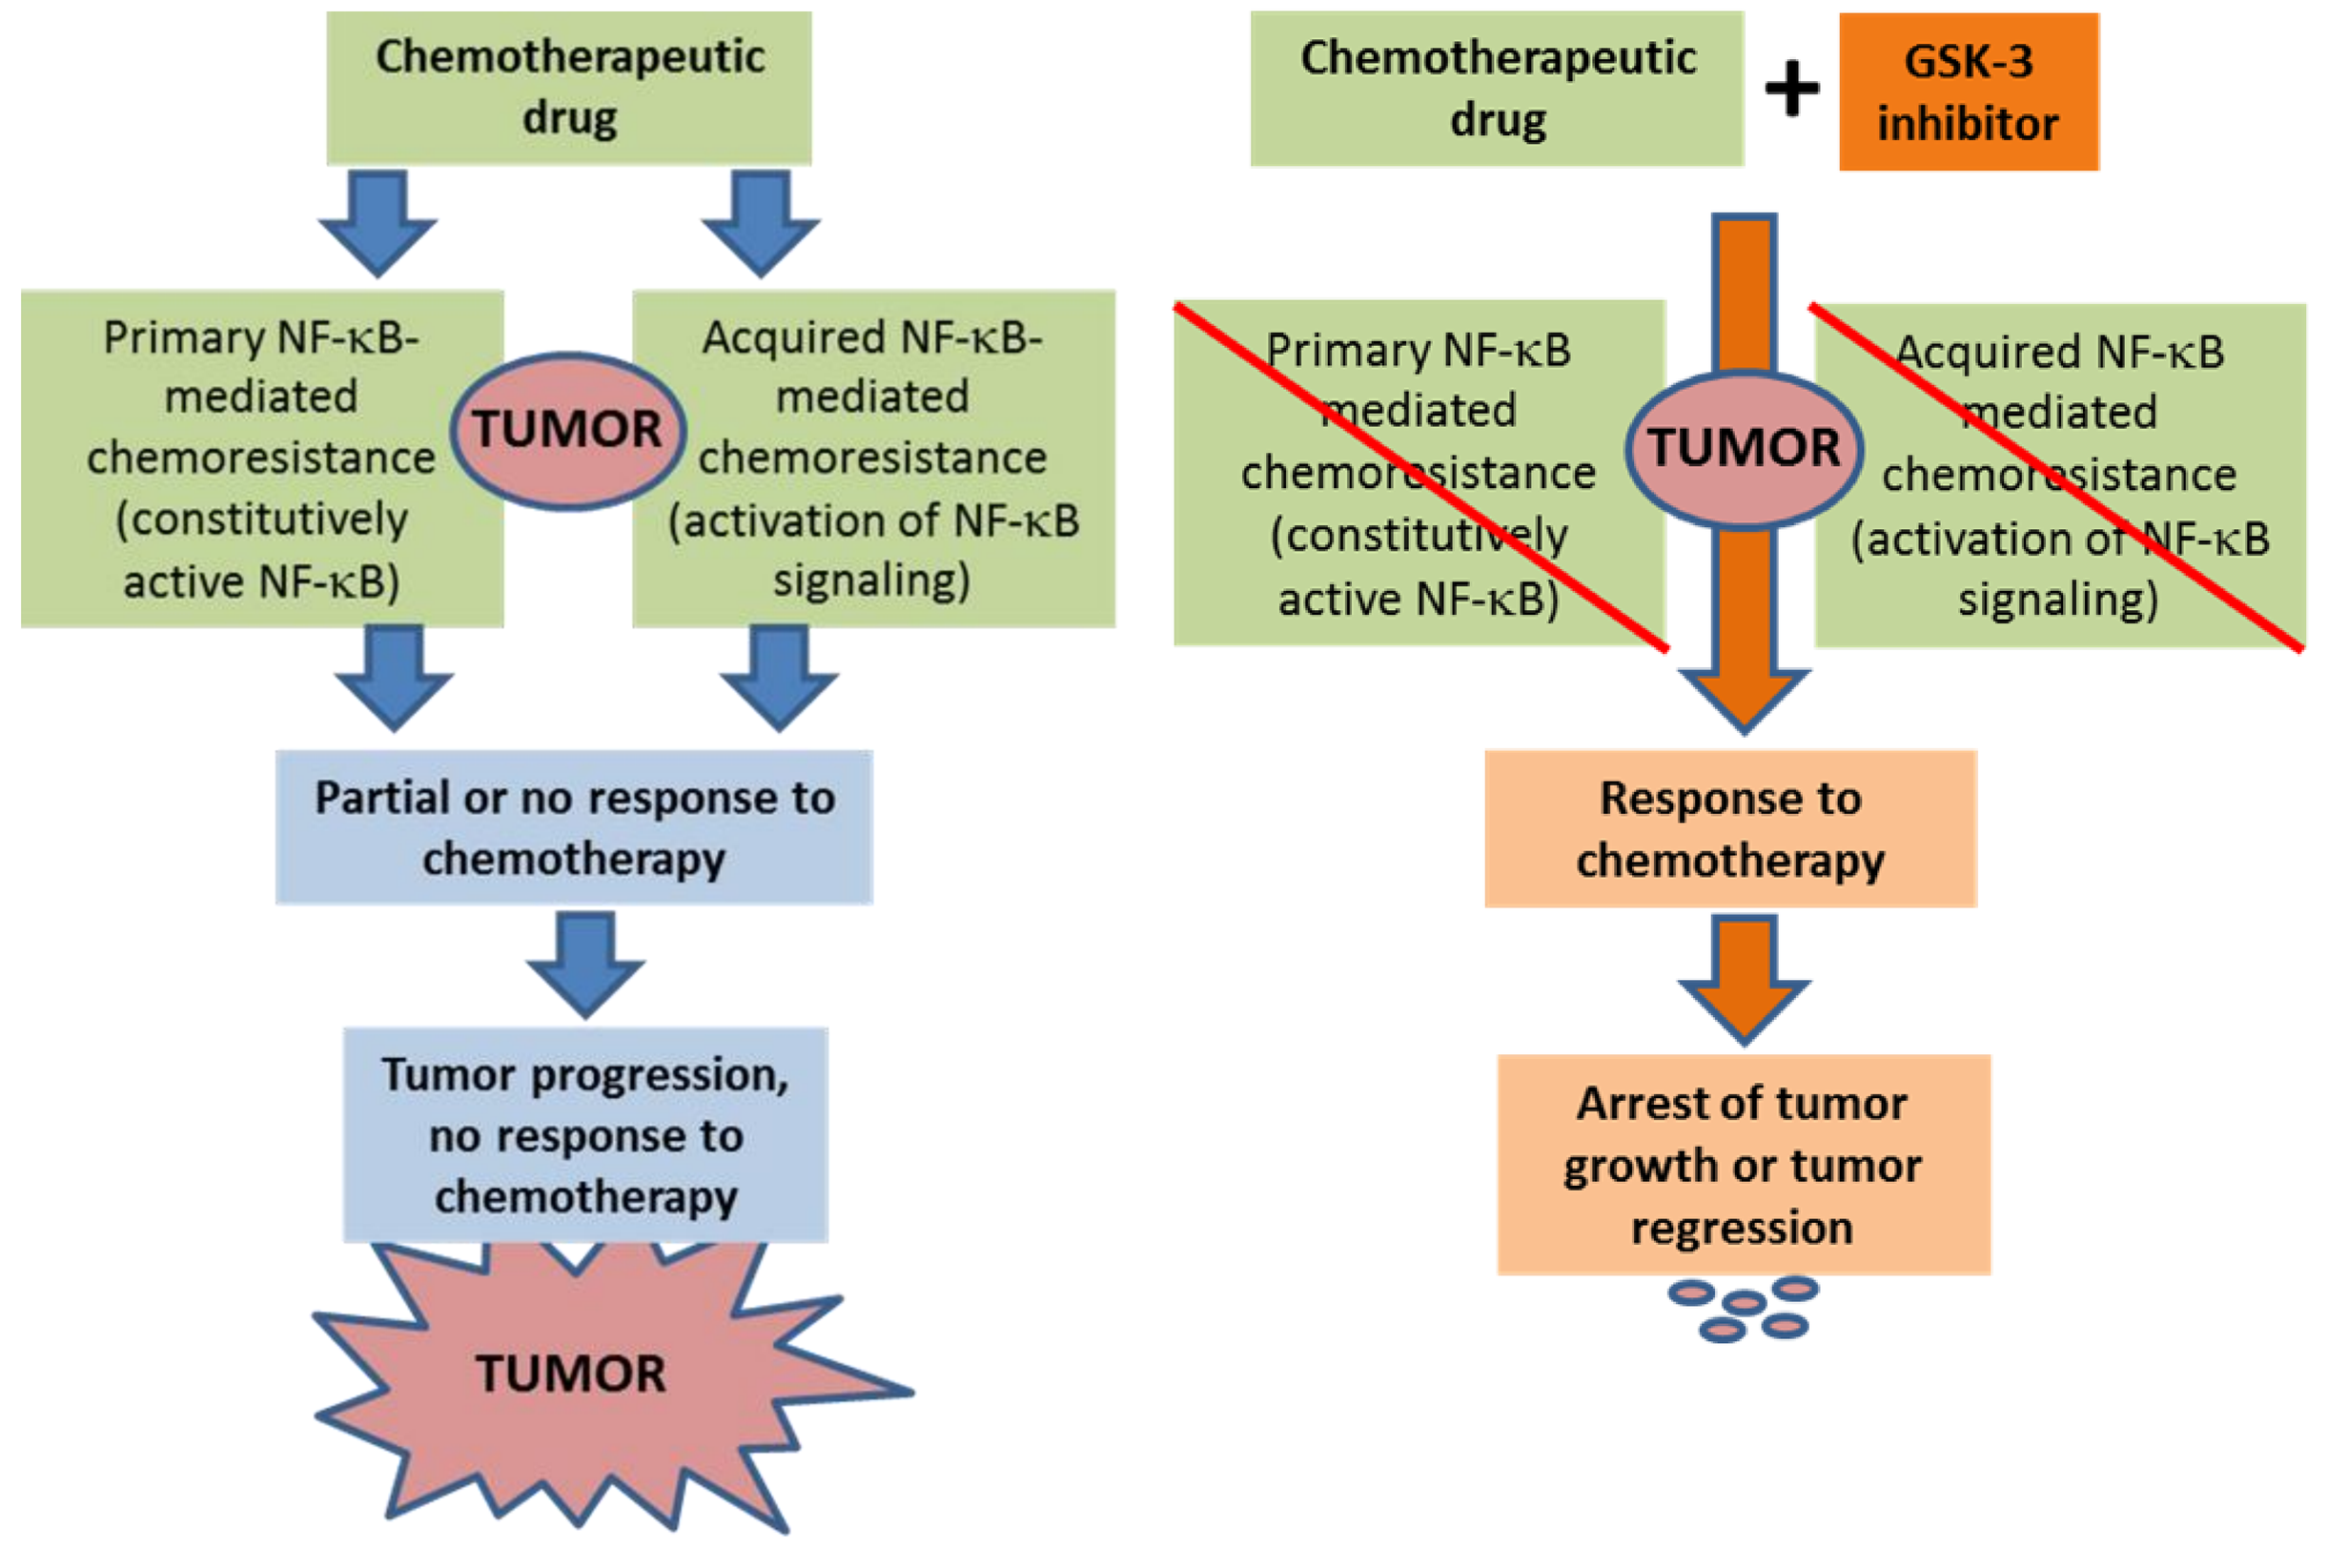

Chemotherapeutic drug
Primary NF-κB-mediated chemoresistance (constitutively active NF-κB)
TUMOR
Acquired NF-κB-mediated chemoresistance (activation of NF-κB signaling)
Partial or no response to chemotherapy
Tumor progression, no response to chemotherapy
TUMOR
Chemotherapeutic drug
+
GSK-3 inhibitor
Primary NF-κB mediated chemoresistance (constitutively active NF-κB)
TUMOR
Acquired NF-κB mediated chemoresistance (activation of NF-κB signaling)
Response to chemotherapy
Arrest of tumor growth or tumor regression

Hypothesis: inhibition of GSK-3β, a positive regulator of NF-κB activity, overcomes NF-κB-mediated chemoresistance and thereby potentiates the effect of conventional chemodrugs in human neuroblastoma.

Results (1)

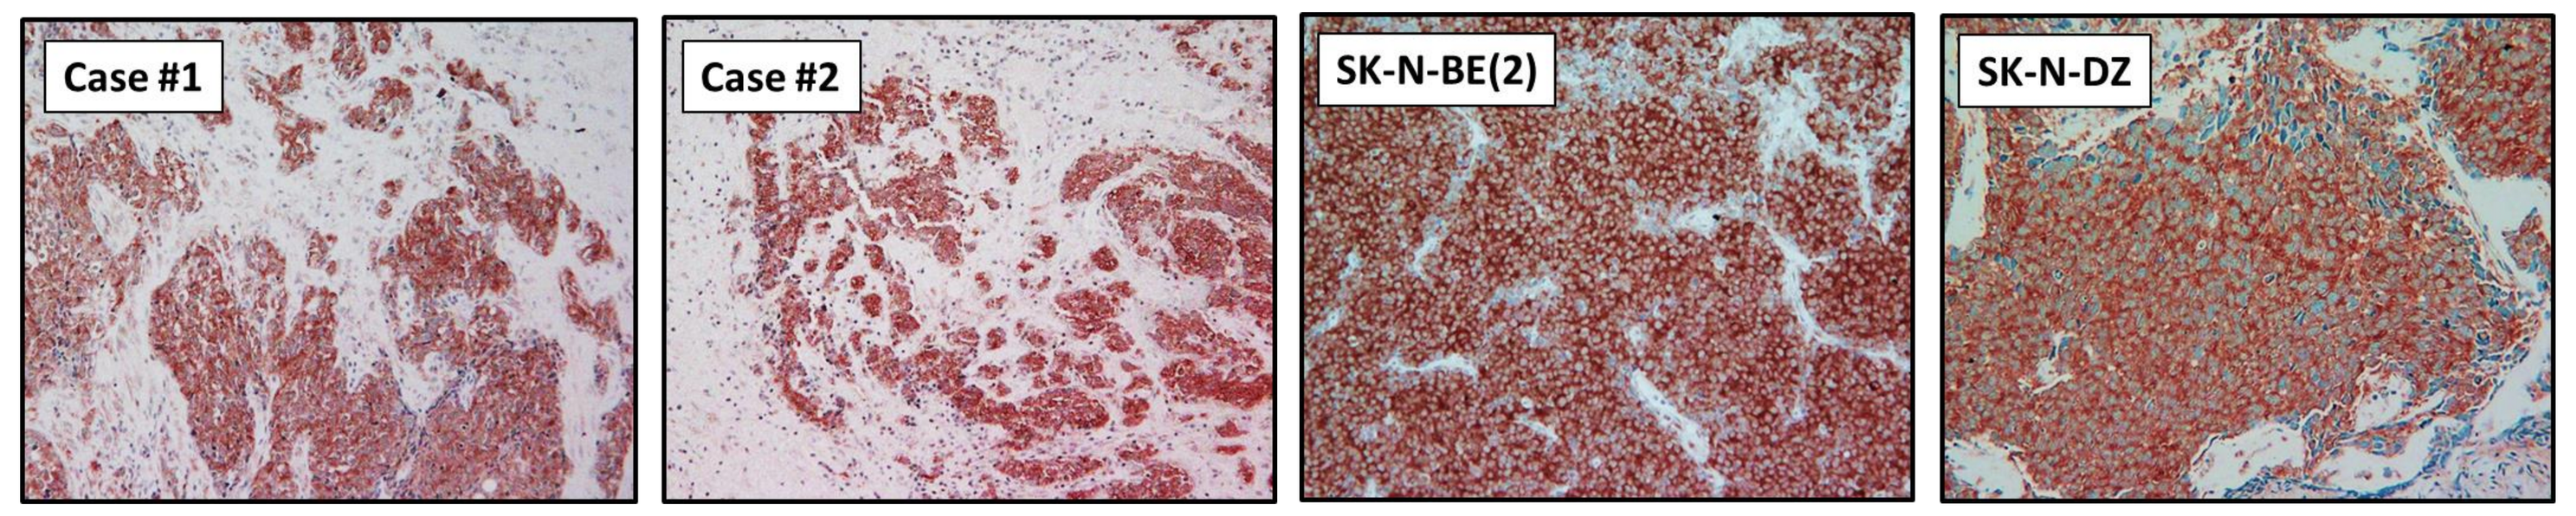

Case #1
Case #2
SK-N-BE(2)
SK-N-DZ

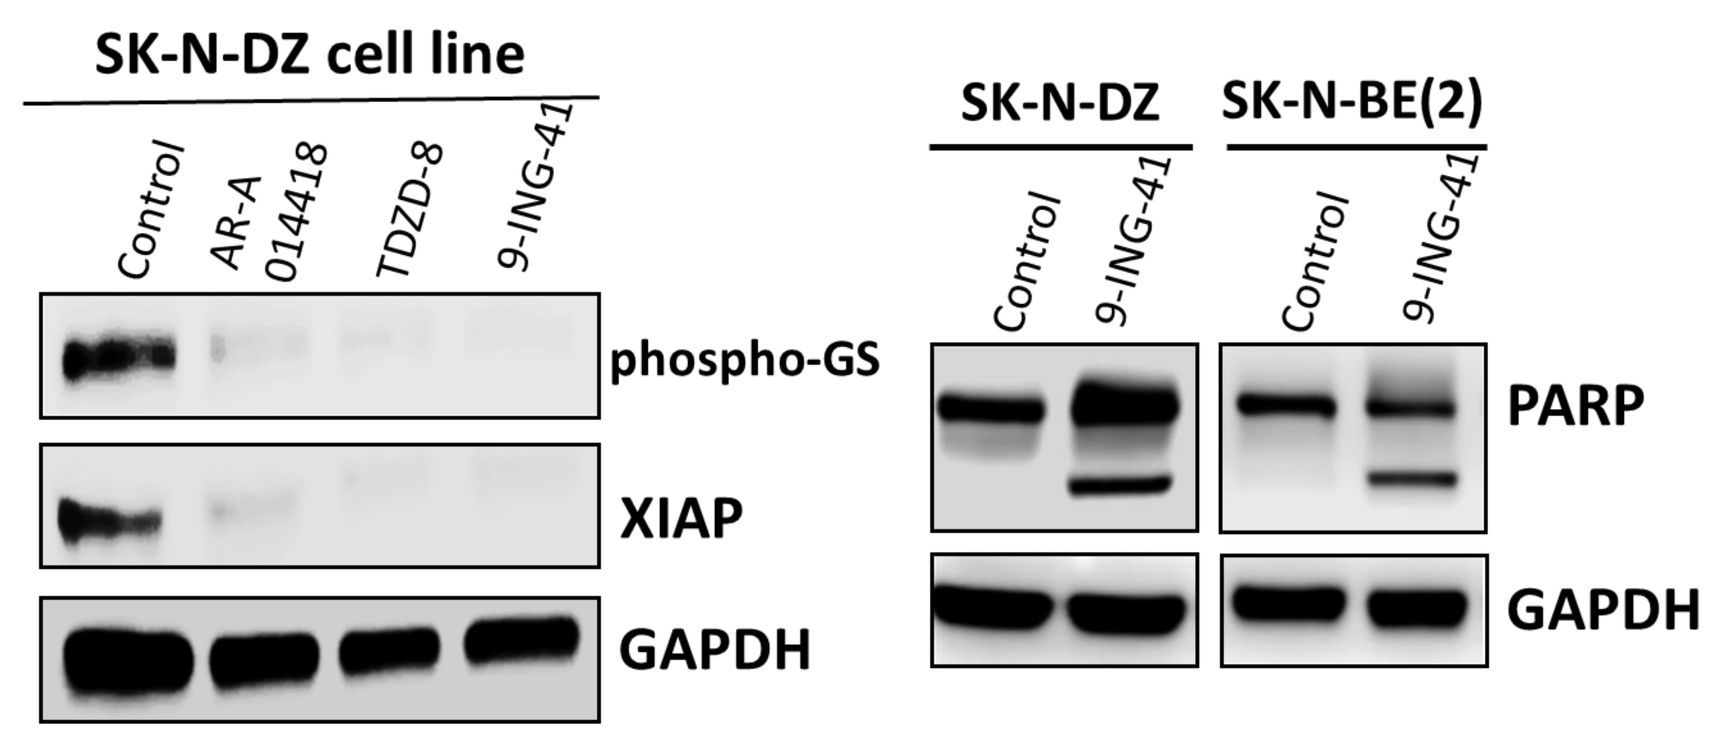

SK-N-DZ cell line
Control
AR-A 014418
TDZD-8
9-ING-41
phospho-GS
XIAP
GAPDH
SK-N-DZ
SK-N-BE(2)
Control
9-ING-41
Control
9-ING-41
PARP
GAPDH

Figure 1. Inhibition of GSK-3 suppresses XIAP expression and leads to apoptosis in neuroblastoma cells. *Upper panel*, immunohistochemical analysis of GSK-3β expression and localization in human neuroblastoma tumors obtained from cancer patients (Case #1 and Case #2) and in xenograft tumors established from SK-N-BE(2) and SK-N-DZ neuroblastoma cell lines. *Lower panel*, neuroblastoma cells were treated with 5 μM AR-A014418, 5 μM TDZD-8 and 0.1 μM (lower left panel) or 1 μM (lower right panel) of 9-ING-41 for 48 hours. 30 μg of proteins (whole cell lysate) was separated by SDS-PAGE, transferred to PVDF membrane, and immunoblotted as indicated.

Using immunohistochemical staining, we found that GSK-3β is highly expressed in human neuroblastomas and in subcutaneous xenograft tumors established from SK-N-BE(2) and SK-N-DZ neuroblastoma cell lines (Fig. 1, upper panel). Using a cell viability assay, we found that the GI_{50} of our novel GSK-3 inhibitor 9-ING-41 (100 nM) for inhibiting the growth of SK-N-DZ and SK-N-BE(2) neuroblastoma cells is more than 80 times lower than the GI_{50} of other GSK-3 inhibitors, namely AR-A014418 and TDZD-8. Our results show that 9-ING-41 inhibits GSK-3, as shown by downregulation of phospho-glycogen synthase (phospho-GS) expression, leading to a decreased expression of NF-κB target XIAP (anti-apoptotic protein), leading to significant apoptosis in neuroblastoma cells *in vitro* as shown by PARP cleavage, an apoptosis marker (Fig. 1, lower panel).

Results (2)

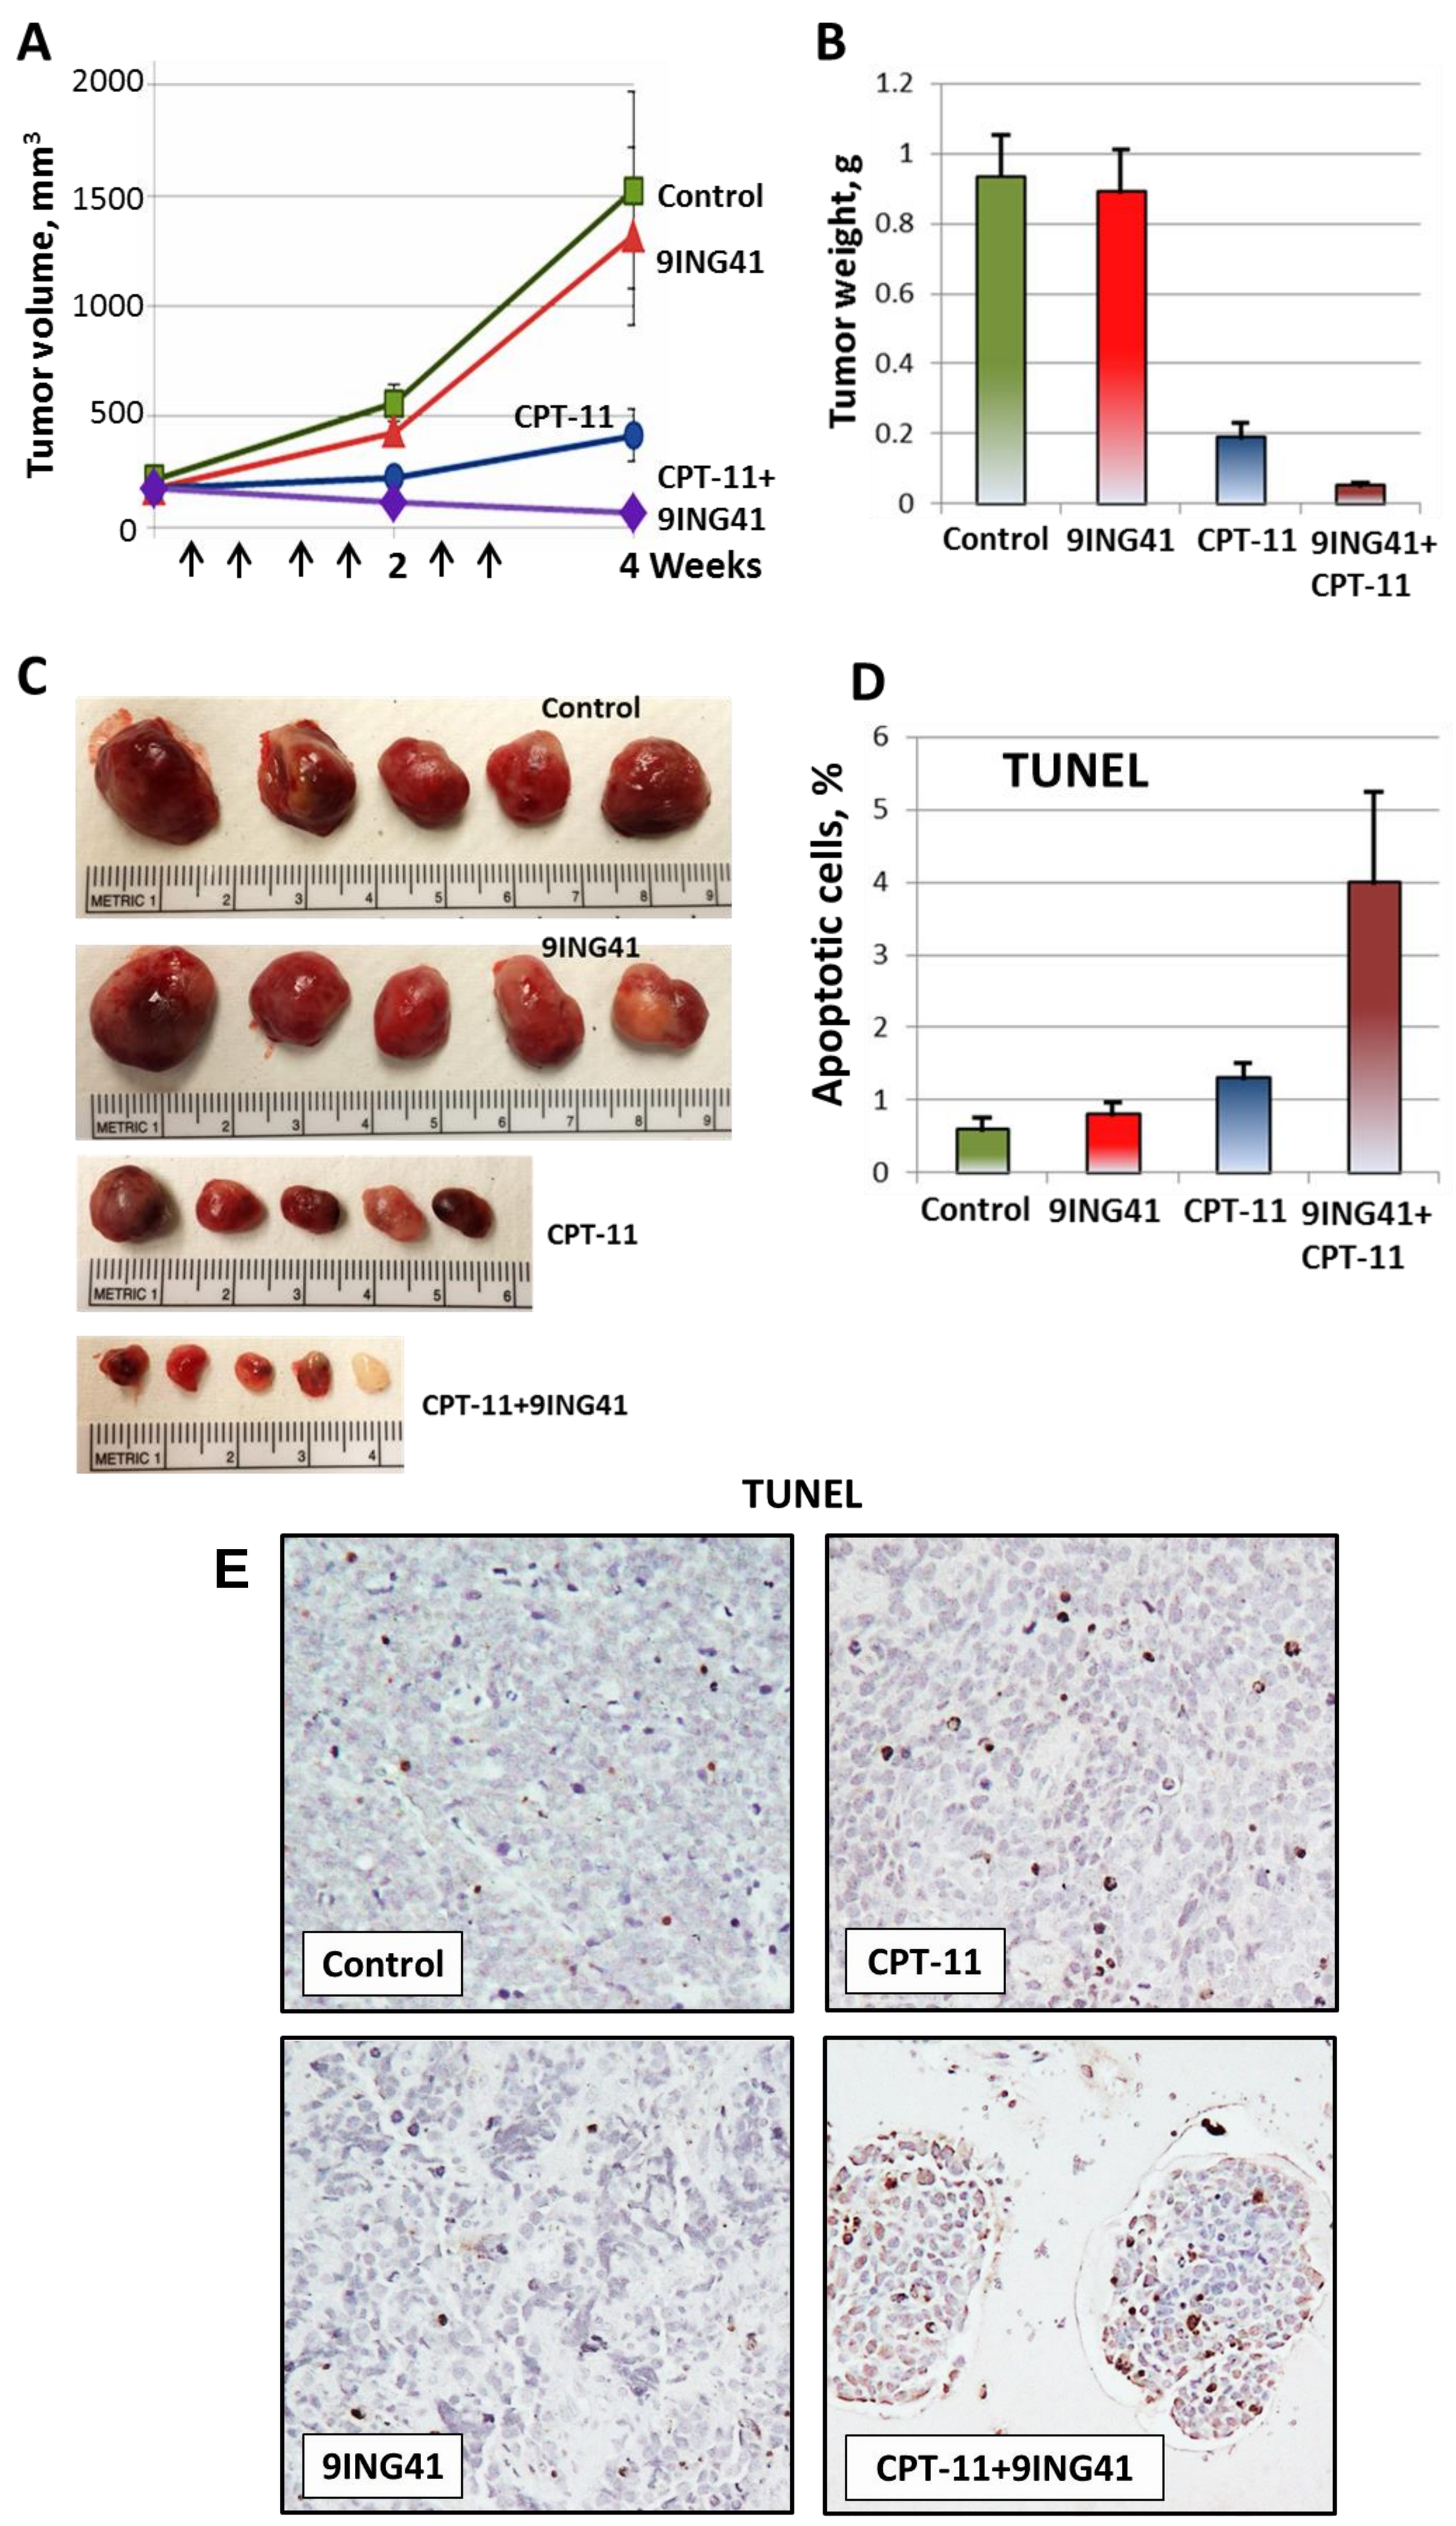

A
Tumor volume, mm3
2000
1500
1000
500
0
Control
9ING41
CPT-11
CPT-11+ 9ING41
2
4 Weeks
B
Tumor weight, g
1.2
1
0.8
0.6
0.4
0.2
0
Control
9ING41
CPT-11
9ING41+ CPT-11
C
Control
9ING41
CPT-11
CPT-11+9ING41
D
TUNEL
Apoptotic cells, %
6
5
4
3
2
1
0
Control
9ING41
CPT-11
9ING41+ CPT-11
TUNEL
E
Control
CPT-11
9ING41
CPT-11+9ING41

Figure 2. Treatment with CPT-11+9ING41 leads to an increased apoptosis and a partial regression of SK-N-DZ xenograft tumors. SK-N-DZ neuroblastoma cells were inoculated subcutaneously (subQ) to 20 nude mice (1 tumor per mouse). Tumors were size matched and mice were randomized into 4 treatment groups: control (DMSO; n=5 mice), CPT-11 (5 mg/kg, n=5 mice), 9ING41 (70 mg/kg, n=5 mice) and Irinotecan+9ING41 (n=5 mice). ***A***, Vehicle (20 μL DMSO) or drugs were injected as shown by arrows. Points, mean tumor volume; bars, SE. ***B***, Mice were sacrificed when tumors grew to more than 5 times the original starting volume and the weight of resected tumors was measured. Columns, mean tumor weight; bars, SE. ***C***, Representative pictures of GBM PDX subQ tumors from each group of animals. ***D***, The percentage of apoptotic cells was determined by TUNEL staining. Columns, mean; bars, SE. ***E***, Representative pictures of TUNEL staining of SK-N-DZ neuroblastoma xenograft tumors treated as indicated.

Conclusion

Our results suggest that the inhibition of GSK-3 is a promising new therapeutic approach to enhance chemosensitivity of neuroblastoma to the antitumor effect of irinotecan.

Materials and Methods

Human neuroblastoma cell lines SK-N-DZ and SK-N-BE(2) were purchased from ATCC. GSK-3 inhibitors AR-A014418 and TDZD-8 and other chemical reagents have been purchased from Sigma-Aldrich. GSK-3β, GAPDH, PARP, phospho-glycogen synthase, Ser641 antibodies were purchased from Cell Signaling Technologies. XIAP antibody has been purchased from BD Biosciences. Cell viability was examined using a colorimetric MTS assay, the CellTiter 96 assay (Promega), according to the manufacturer's protocol. IHC staining was performed with Dako Envision+/HRP kit according to the recommended manufacturer procedure.

References

1. Aggarwal B. Nuclear factor-kB: the enemy within. Cancer Cell 2004;6(3):203–208.
2. Tas S, Vervoordeldonk M, Tak P. Gene therapy targeting nuclear factor-kB: towards clinical application in inflammatory diseases and cancer. Curr Gene Ther 2009;9:160–170.
3. Hoeflich K, Luo J, Rubie E, Tsao M, Jin O, Woodgett J. Requirement for glycogen synthase kinase-3β in cell survival and NF-κB activation. Nature 2000;406:86–90.
4. Ougolkov A, Fernandez-Zapico M, Savoy D, Urrutia R, Billadeau D. Glycogen synthase kinase-3beta participates in nuclear factor kappaB-mediated gene transcription and cell survival in pancreatic cancer cells. Cancer Res 2005;65:2076-2081.
5. Ougolkov A, Fernandez-Zapico M, Bilim V, Smyrk T, Chari S, Billadeau D. Aberrant nuclear accumulation of glycogen synthase kinase-3beta in human pancreatic cancer: association with kinase activity and tumor dedifferentiation. Clin Cancer Res 2006;12:5074-5081.
6. Ougolkov A, Bone N, Fernandez-Zapico M, Kay N, Billadeau D. Inhibition of glycogen synthase kinase-3 activity leads to epigenetic silencing of nuclear factor kappaB target genes and induction of apoptosis in chronic lymphocytic leukemia B cells. Blood 2007;110:735-742.
7. Ougolkov A, Shakoori A, Zhang B, Modarressi M, Billadeau D, Mai M, et al. Deregulated GSK3beta activity in colorectal cancer: its association with tumor cell survival and proliferation. Biochem Biophys Res Commun 2005;334:1365-1373.
8. Bilim V, Ougolkov A, Yuuki K, Naito S, Kawazoe H, Muto A, et al. Glycogen synthase kinase-3: a new therapeutic target in renal cell carcinoma. Br J Cancer 2009;101:2005-2014.
9. Naito S, Bilim V, Yuuki K, Ugolkov A, Motoyama T, Nagaoka A, et al. Glycogen synthase kinase-3beta: a prognostic marker and a potential therapeutic target in human bladder cancer. Clin Cancer Res 2010;16:5124-5132.
10. Ammann JU, Haag C, Kasperczyk H, Debatin KM, Fulda S. Sensitization of neuroblastoma cells for TRAIL-induced apoptosis by NF-kappaB inhibition. Int J Cancer 2009;124(6):1301-1311.
11. Shimasaki T, Ishigaki Y, Nakamura Y, Takata T, Nakaya N, Nakajima H, et al. Glycogen synthase kinase- 3β inhibition sensitizes pancreatic cancer cells to gemcitabine. J Gastroenterol. 2012;47:321-333.
12. Dickey A, Schleicher S, Leahy K, Hu R, Hallahan D, Thotala D. GSK-3β inhibition promotes cell death, apoptosis, and in vivo tumor growth delay in neuroblastoma Neuro-2A cell line. J Neurooncol. 2011;104:145-153.
13. Carter Y, Kunnimalaiyaan S, Chen H, Gamblin T, Kunnimalaiyaan M. Specific glycogen synthase kinase-3 inhibition reduces neuroendocrine markers and suppresses neuroblastoma cell growth. Cancer Biol Ther. 2014;15:510-515.